Discovery of TSC-100: A Natural HA-1-specific TCR to Treat Leukemia Following Hematopoietic Stem Cell Transplant Therapy

Ribhu Nayar, Sonal Jangalwe, Mollie M. Jurewicz, Antoine J. Boudot, Andrew S. Basinski, Robert Prenovitz, Elizabeth A. Olesin, Daniel C. Pollacksmith, Qikai Xu, Yifan Wang, Amy Virbasius, Jeffrey Li, Holly Whitton, Garrett S. Dunlap, Alexander Cristofaro, Nancy Nabilsi, Ruan Zhang, Candace Perullo, Sida Liao, Kenneth L. Jahan, Kenneth Olivier, Gavin MacBeath

#156

Abstract

Background: Approximately 30-40% of AML patients relapse following allogeneic hematopoietic stem cell transplant therapy, leaving them with very few treatment options (1,2). Rare patients that naturally develop an HA-1-specific graft-versus-leukemia T cell response, however, show substantially lower relapse rates (3,4). HA-1 (VL**H**DDLLEA, genotype RS_1801284 A/G or A/A) is an HLA-A*02:01- and haematopoietically-restricted minor histocompatibility antigen, making it an ideal candidate for TCR immunotherapy for liquid tumors (5).

Methods: We developed a high-throughput TCR discovery platform that enables rapid cloning of antigen-specific TCRs from healthy donors. We then used this platform to screen 178.3 million naïve CD8+ T cells from six unique HA-1- (VL**R**DDLLEA, genotype RS_1801284 G/G) donors, identifying 329 HA-1-specific TCRs. We tested each TCR for expression and the ability to kill HA-1+ target cells, using a previously published, clinical-stage HA-1-specific TCR as a benchmark for these studies (6). In parallel, we tested TCR constant region modifications to promote expression and proper pairing of exogenous TCR alpha and beta chains and designed a lentiviral vector to co-deliver CD8 coreceptors as well as a CD34 enrichment tag to enable purification of engineered T cells. The top 11 candidates were cloned into our optimized backbone and evaluated for cytotoxicity, cytokine production, and T cell proliferation using a panel of HLA-A*02:01+ HA-1+ cell lines. Finally, the top two TCRs were evaluated for alloreactivity and off-target cross-reactivity using our proprietary genome-wide T-Scan platform.

Results: The TCR discovery and evaluation platform described here identified 329 HA-1-specific TCRs from a total of 178.3 million naïve T cells, and TSC-100 as the most active TCR. Defined mutations in the constant region of TSC-100 enhanced its surface expression while decreasing expression of endogenous TCRs, and co-introduction of CD8 enabled efficient engagement and function of engineered CD4 T cells. Overall, TSC-100 exhibited comparable activity to a clinical-stage benchmark TCR when challenged with cell lines expressing moderate to high levels of HA-1, and superior activity when incubated with cell line expressing low levels of both HA-1 and MHC-I (6). In addition, TSC-100 exhibited no detectable allo-reactivity to 108 different HLA types tested, and no appreciable off-target effects when challenged with a genome-wide library expressing peptides derived from human proteins.

Conclusion: TSC-100 exhibits comparable or superior activity to a clinical-stage therapeutic TCR, with no appreciable allo-reactivity or off-target effects. Based on these results, TSC-100 has been advanced to IND-enabling activities to prepare for first-in-human testing in 2021.

TCR discovery platform identifies 329 novel HA-1 TCRs

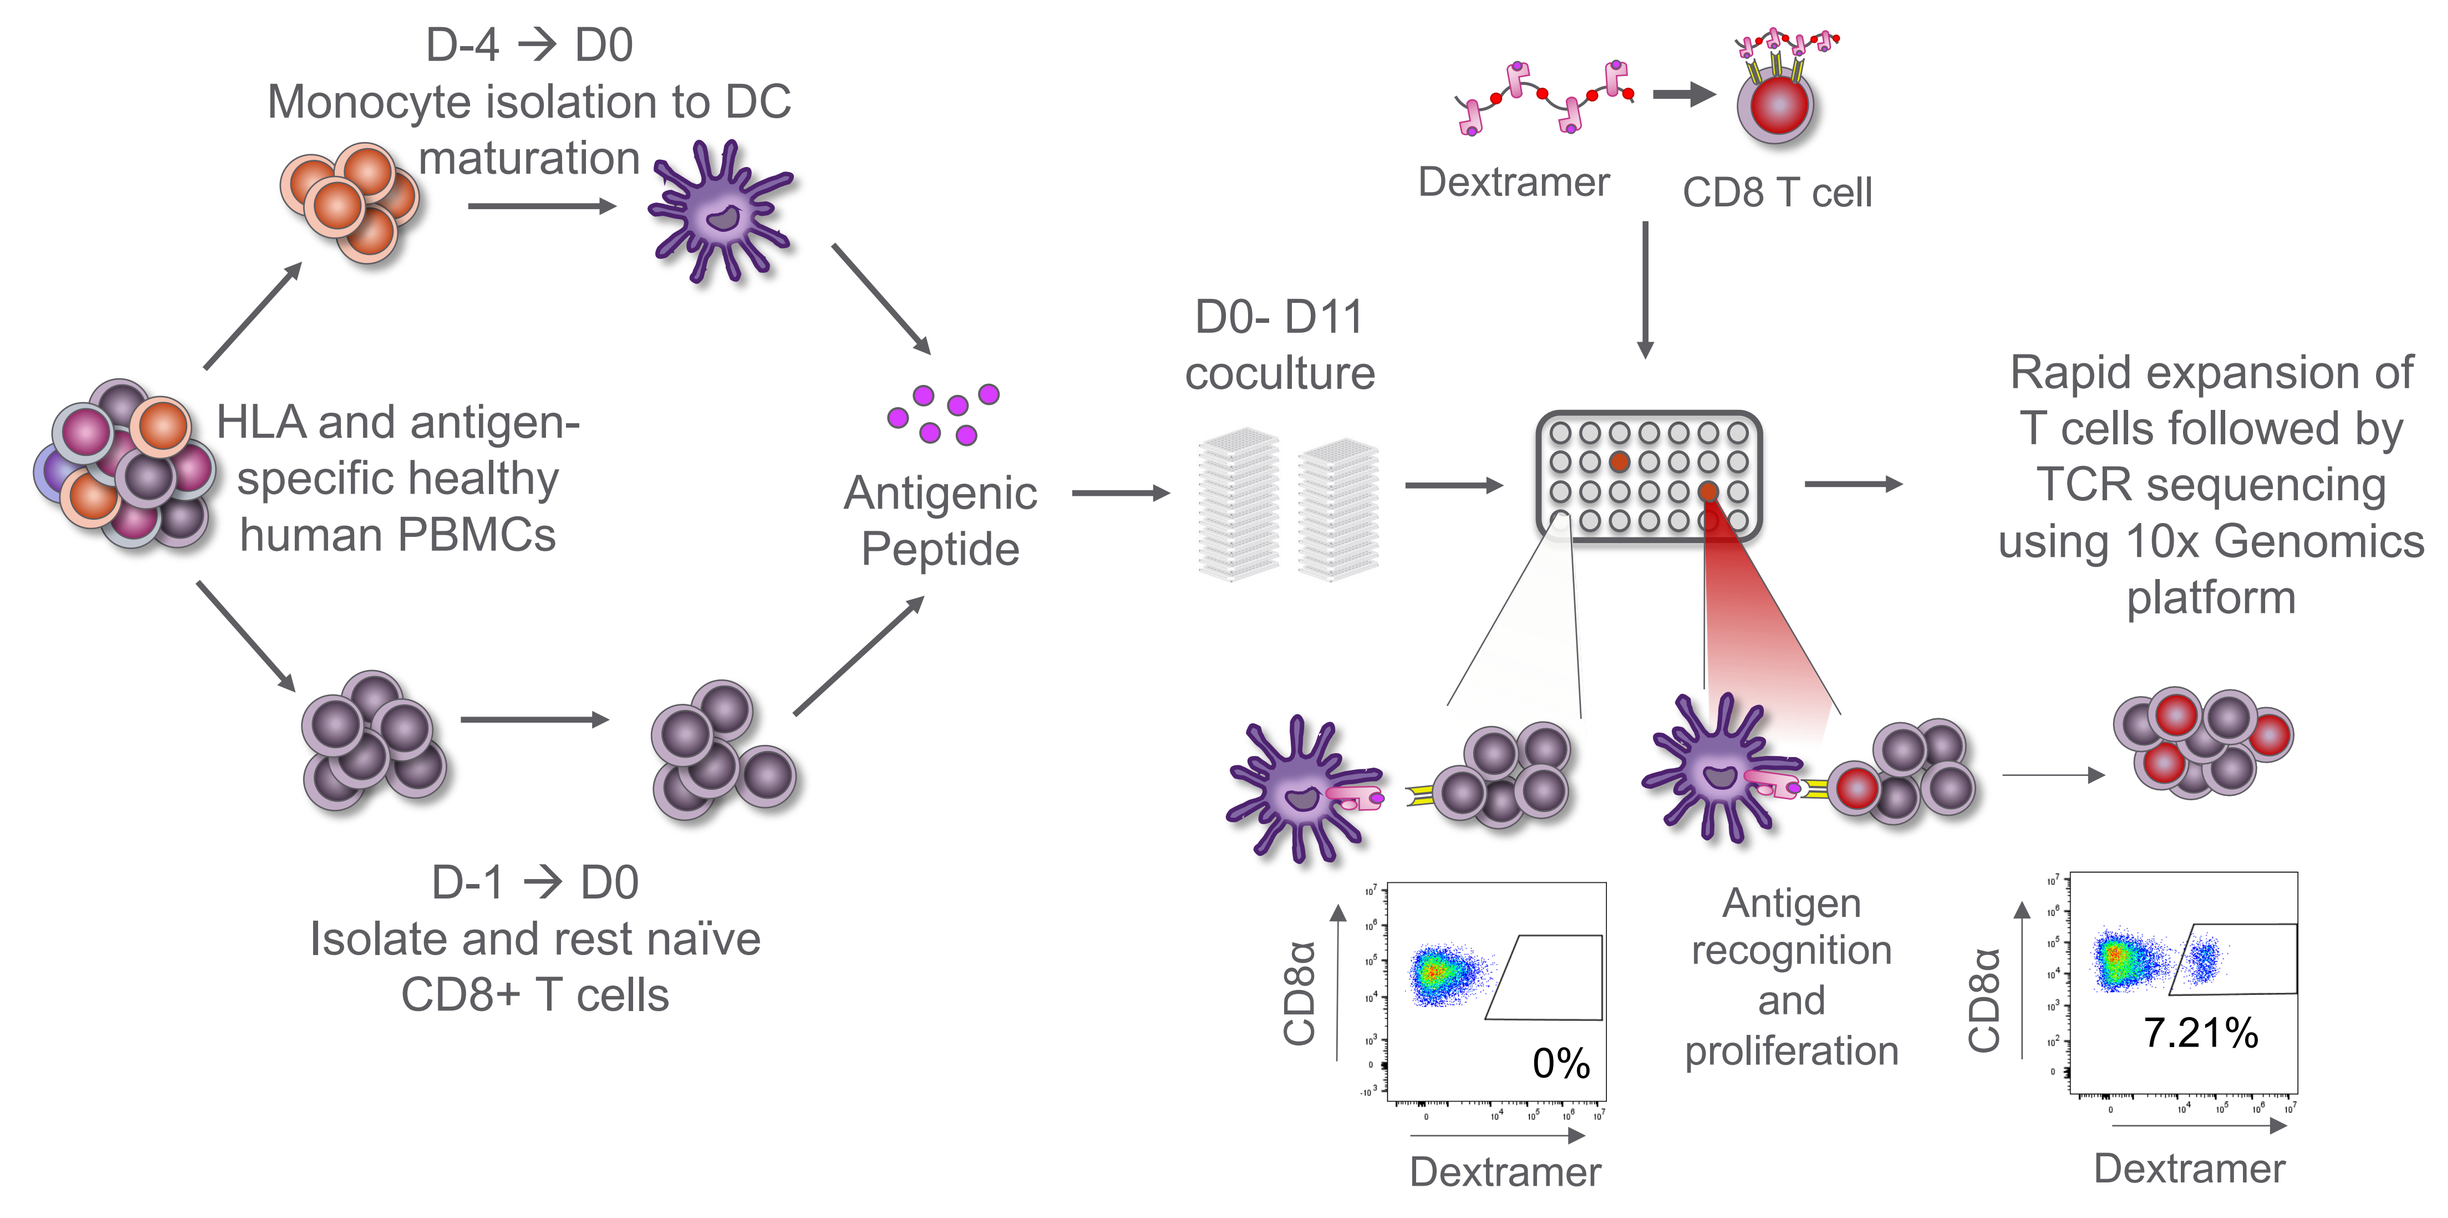
D-4 → D0
Monocyte isolation to DC maturation
HLA and antigen-specific healthy human PBMCs
Antigenic Peptide
D-1 → D0
Isolate and rest naïve CD8+ T cells
Dextramer
CD8 T cell
D0- D11 coculture
Rapid expansion of T cells followed by TCR sequencing using 10x Genomics platform
Antigen recognition and proliferation
CD8α
0%
7.21%
Dextramer

CD14+ monocytes were isolated from PBMCs of six HA-1-negative healthy donors on day -4 and differentiated to mature DCs. On day -1, naïve CD8 T cells were isolated from autologous PBMCs and rested overnight. Co-culture of CD8 T cells and DCs was performed following 3 h pulsing of DCs with 1 µg/mL HA-1 peptide (VL**H**DDLLEA), followed by an 11-day cell expansion phase. Dextramer staining was performed with A*02:01-specific HA-1 dextramer to identify clones, which were then rapidly expanded using irradiated unmatched PBMCs in the presence of IL-2. After 7 days, HA-1 dextramer-positive cells were sorted and single cell TCR sequencing was performed.

"Validate As You Go" (VAYG) screen identifies 11 TCRs with high expression and cytotoxicity

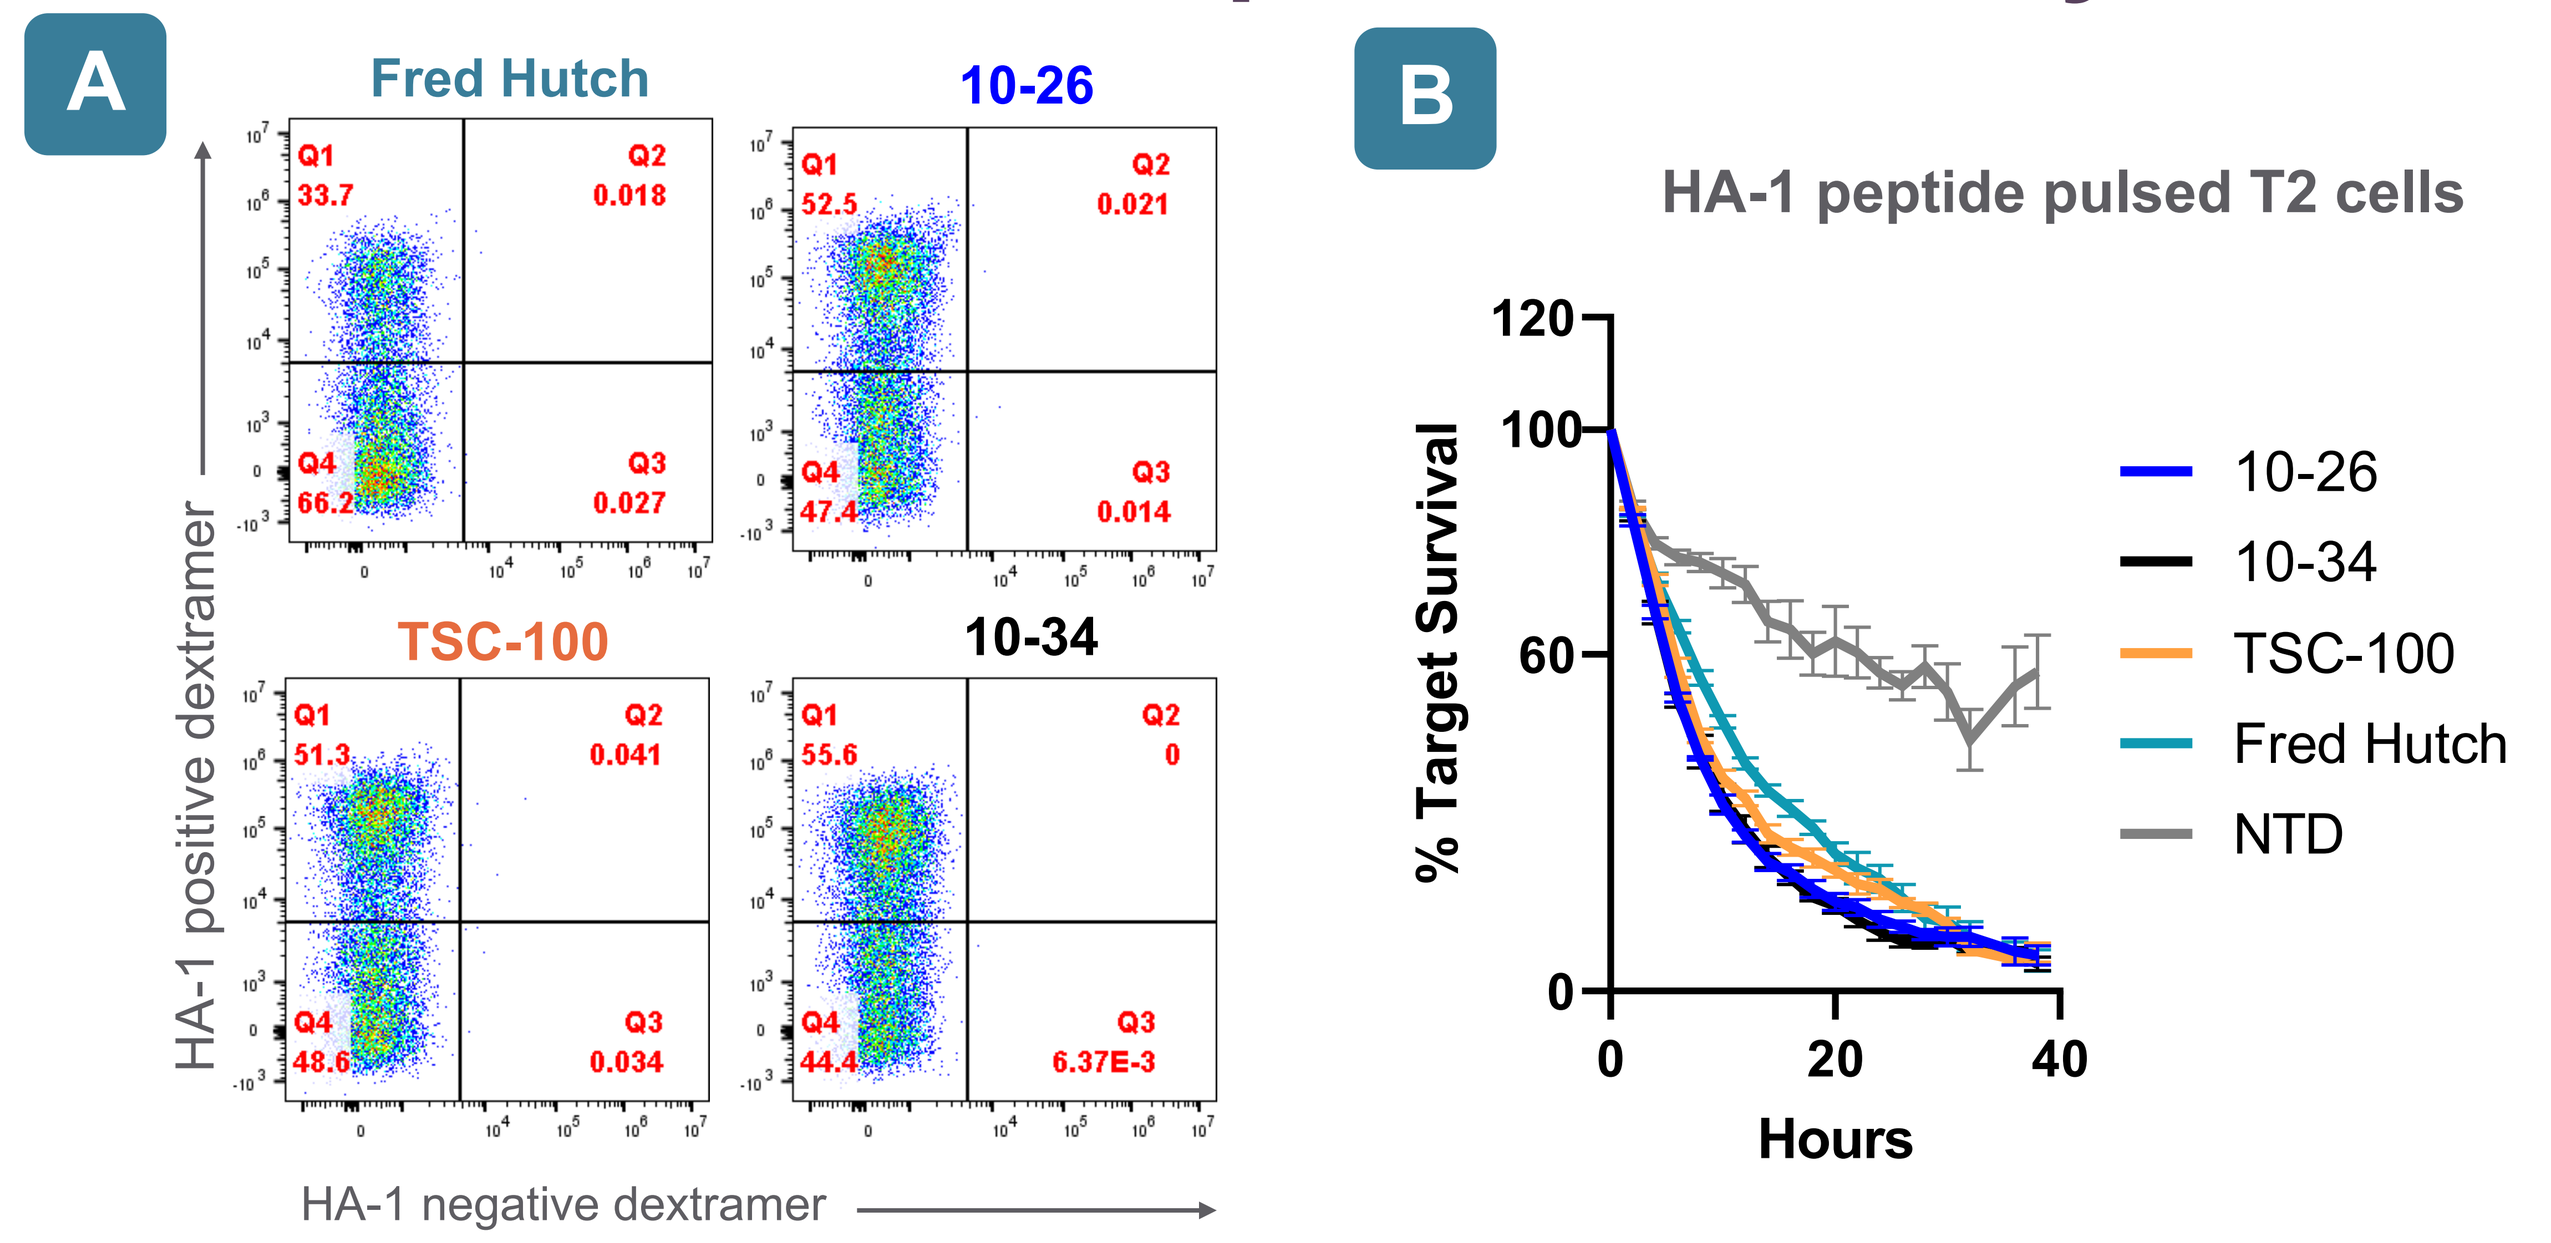
A
Fred Hutch
10-26
TSC-100
10-34
HA-1 positive dextramer
HA-1 negative dextramer
B
HA-1 peptide pulsed T2 cells
% Target Survival
Hours
10-26
10-34
TSC-100
Fred Hutch
NTD

(A) Pan-T cells were transduced to express 329 HA-1-specific TCRs. Surface expression of the TCRs was assessed by HA-1/HLA-A*02:01 dextramer staining. Three of the high-expressing TCRs along with the benchmark 'Fred Hutch' TCR are shown. (B) Engineered T cells were co-cultured with labeled T2 cells loaded with 1 ng/mL HA-1 peptide (E:T=5:1) and survival of the T2 cells was quantified by time-dependent imaging as a readout of T cell cytotoxicity. Non-transduced cells (NTD) served as control.

TScan's optimal vector includes CD8α and CD8β, a CD34 enrichment tag, and modifications in the constant and transmembrane domains of the HA-1 TCR

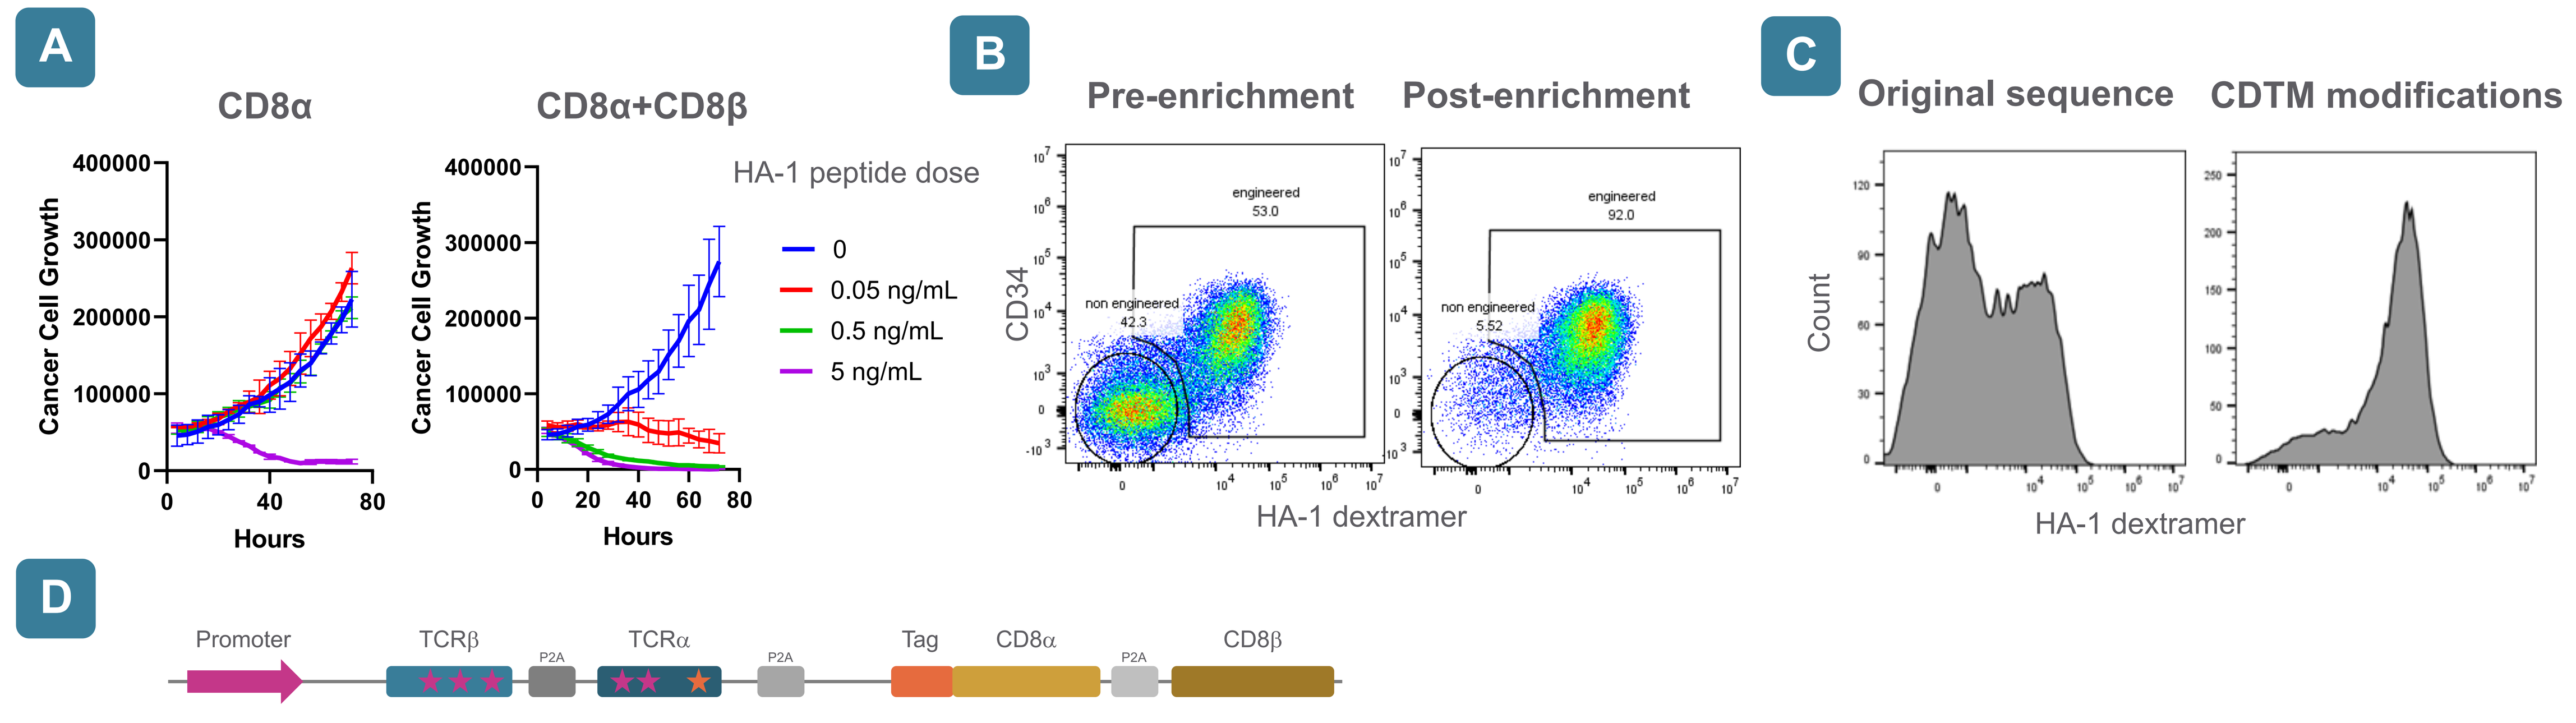
A
CD8α
CD8α+CD8β
HA-1 peptide dose
0
0.05 ng/mL
0.5 ng/mL
5 ng/mL
Cancer Cell Growth
Hours
B
Pre-enrichment
Post-enrichment
engineered
53.0
92.0
non engineered
42.3
5.52
CD34
HA-1 dextramer
C
Original sequence
CDTM modifications
Count
HA-1 dextramer
D
Promoter
TCRβ
P2A
TCRα
Tag
CD8α
CD8β

(A) CD4 T cells were isolated and transduced to express a HA-1-specific TCR along with either CD8α alone or both CD8α and CD8β. Engineered T cells were co-cultured with labeled T2 cells loaded with different concentrations of HA-1 peptide (E:T=5:1) and the growth of the T2 cells (or lack thereof) was quantified on an IncuCyte as a readout of the cytotoxicity of the T cells. (B) TSC-100 delivery vector contains a CD34 tag grafted on the N terminus of CD8α, enabling enrichment of engineered T cells using a GMP-compatible Miltenyi CD34 enrichment kit. T cells were transduced with TSC-100 and evaluated for CD34 expression and TCR expression by HA-1/HLA-A*02:01 dextramer staining pre- and post-CD34 enrichment. (C) The original sequence of TSC-100 was compared to modified sequences carrying optimization of residues in the constant domain (CD) and in the transmembrane domain (TM) designed to ensure surface expression and proper alpha/beta pairing of the TCR. Primary human T cells were transduced with different versions of the TCRs and the surface expression of the properly paired TCR was assessed by dextramer staining. (D) Final vector configuration. Stars indicate CD and TM modifications.

Final triage platform identifies HA-1 TCR TSC-100

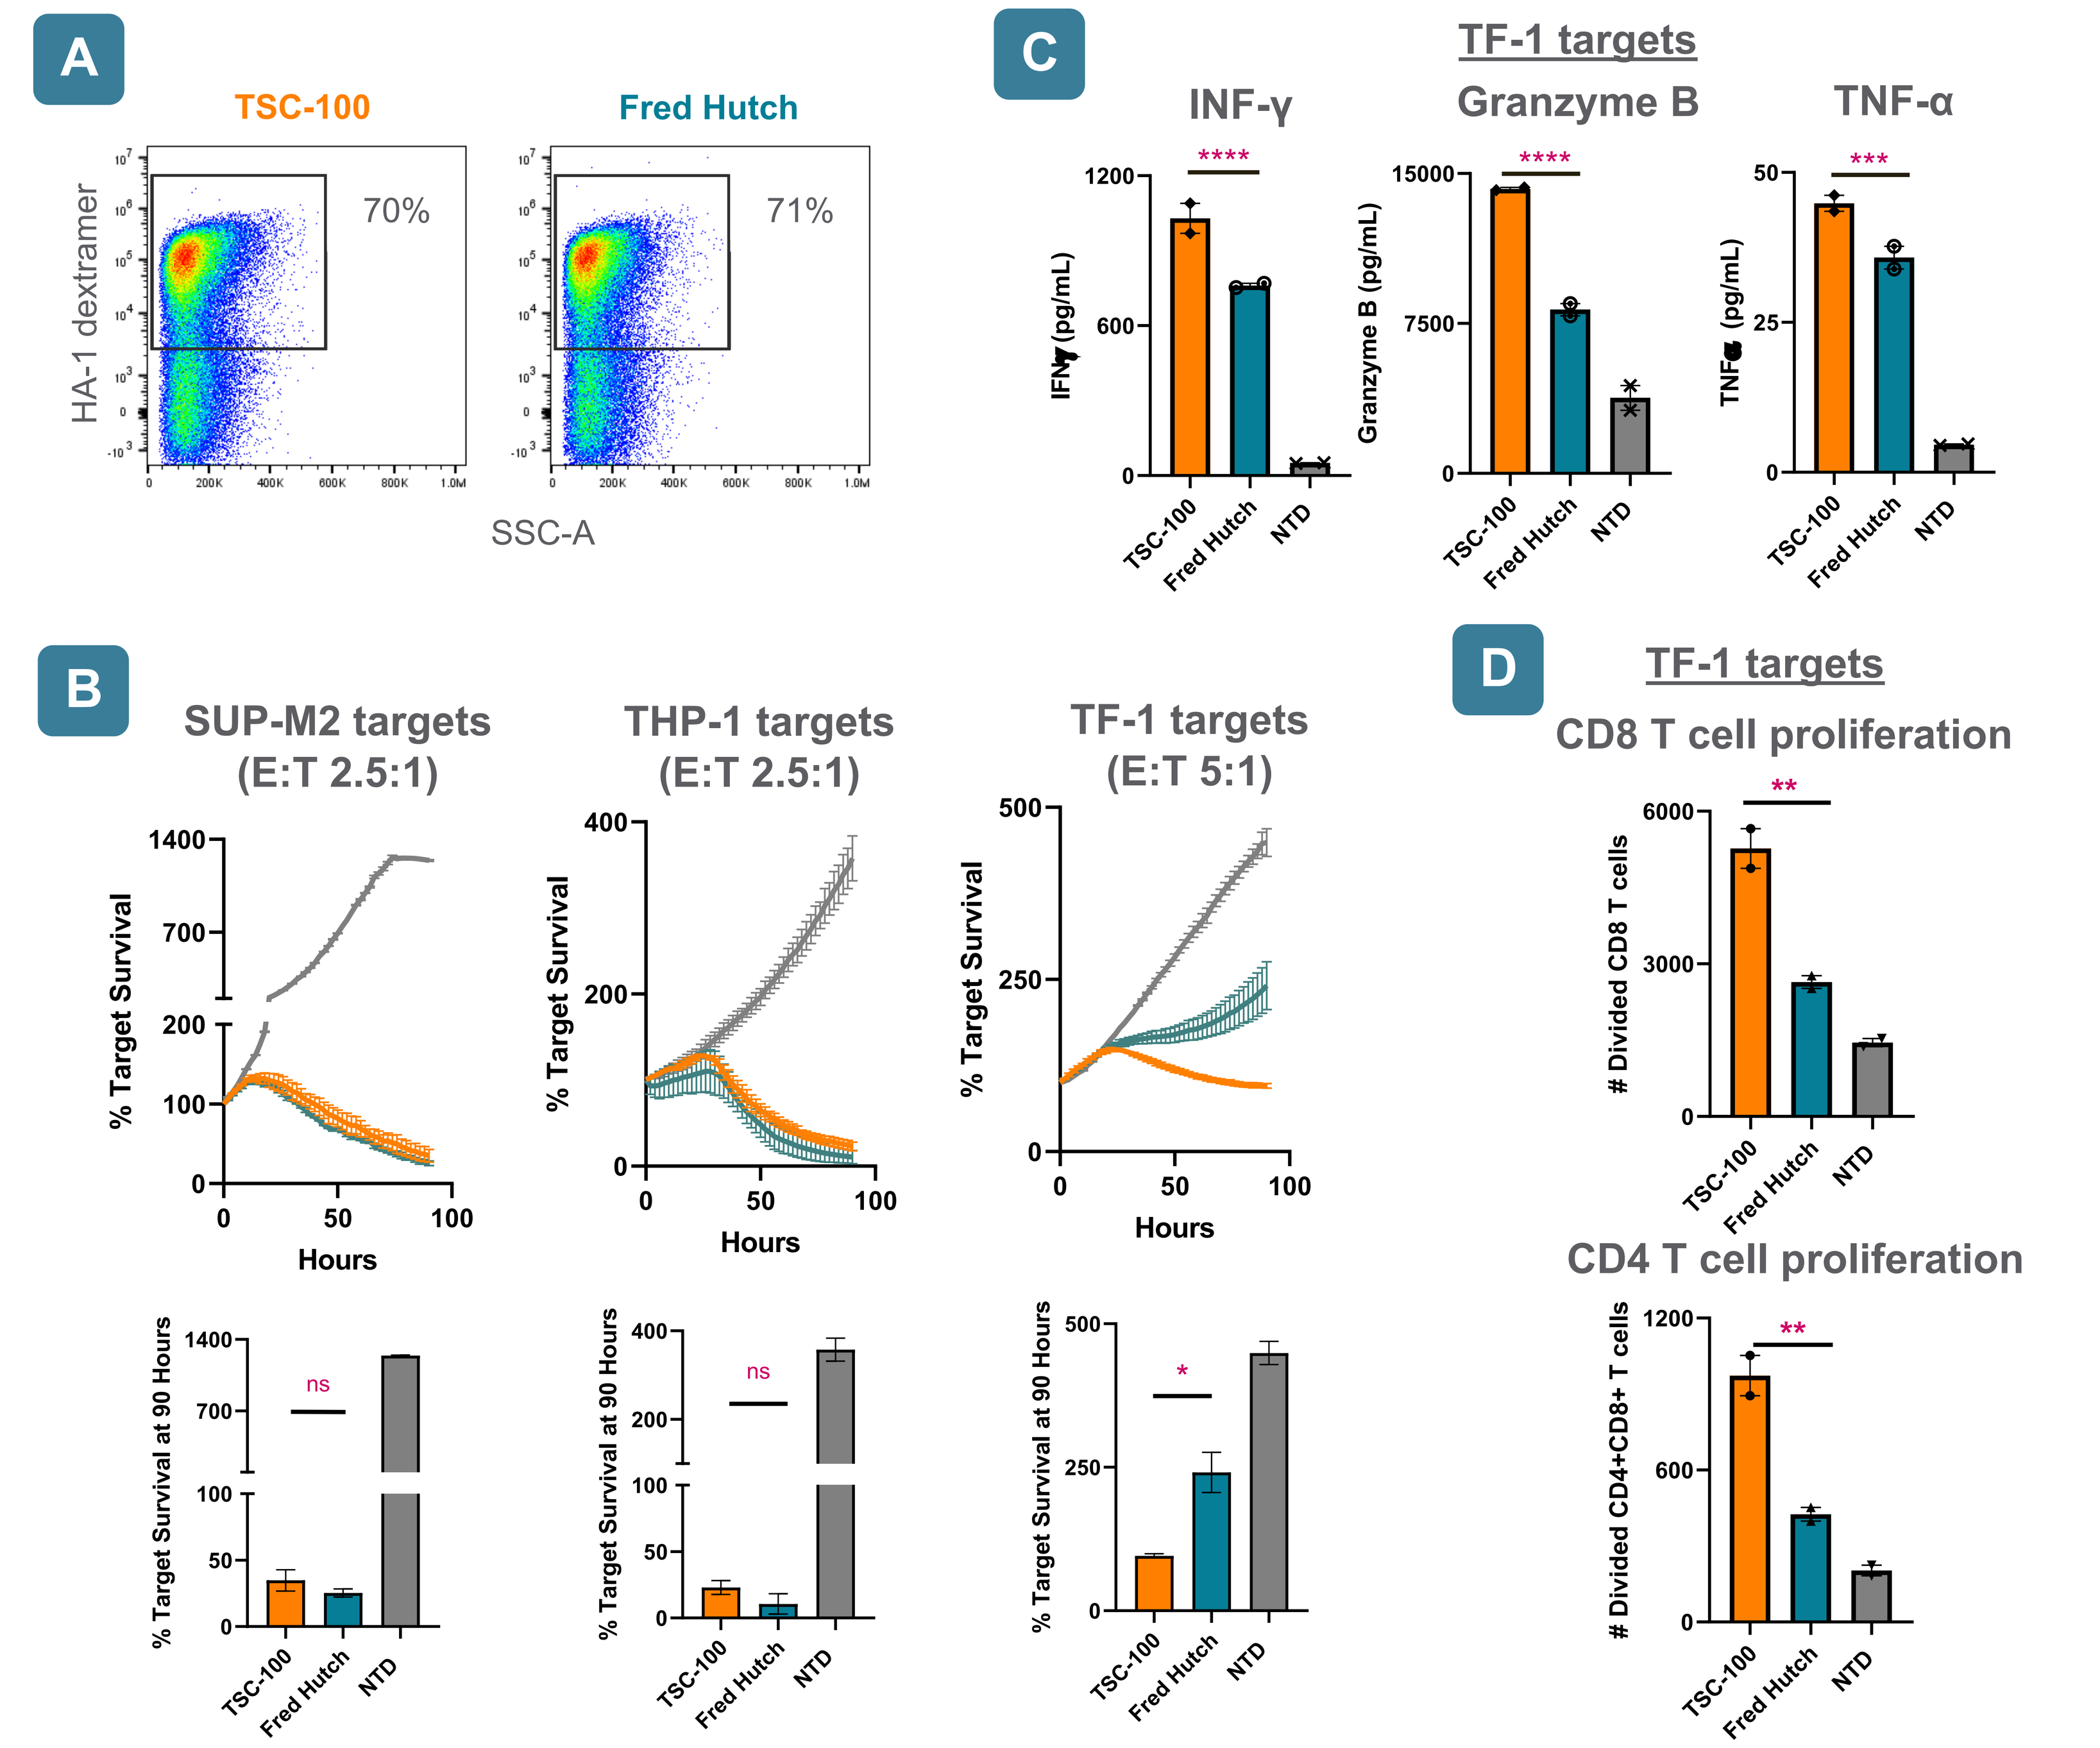
A
TSC-100
Fred Hutch
70%
71%
HA-1 dextramer
SSC-A
B
SUP-M2 targets (E:T 2.5:1)
THP-1 targets (E:T 2.5:1)
TF-1 targets (E:T 5:1)
% Target Survival
Hours
% Target Survival at 90 Hours
ns
*
C
TF-1 targets
INF-γ
Granzyme B
TNF-α
IFNγ (pg/mL)
Granzyme B (pg/mL)
TNFα (pg/mL)
D
TF-1 targets
CD8 T cell proliferation
Divided CD8 T cells
CD4 T cell proliferation
Divided CD4+CD8+ T cells
TSC-100
Fred Hutch
NTD

(A) Pan-T cells were transduced to express 11 HA-1-specific TCRs in the TScan optimal vector. Surface expression of the TCRs was assessed by HA-1/HLA-A*02:01 dextramer staining. TSC-100 and the benchmark TCR 'Fred Hutch' are shown. (B) Engineered T cells were co-cultured with labeled HLA-A*02:01+ cell lines endogenously expressing varying levels of HA-1 at different E:T ratios, and their survival was quantified on an IncuCyte as a readout of cytotoxicity of the T cells. (C) Engineered T cells were co-cultured with these cell lines for 24 h (E:T 1:1) and the concentration of IFN-γ, granzyme B, and TNF-α in the supernatants was determined. Levels of cytokines are shown for TF-1 cells expressing low level of MHC-I and antigen. (D) Engineered T cells were labeled with a proliferation dye and cocultured with target cell lines for 96 h (E:T 1:1). Dye dilution was used to assess proliferation of CD8 and CD4 T cells. Counting beads were added to the samples prior to analysis by flow cytometry and absolute numbers of divided CD8 and CD4 T cells were determined. Proliferation is shown for TF-1 target cells. TCRs were compared by one-way ANOVA followed by Dunnett's multiple comparison test. * P<0.05, ** P<0.005, *** P<0.0005, **** P<0.0001

TSC-100 shows no alloreactivity to 108 HLA types

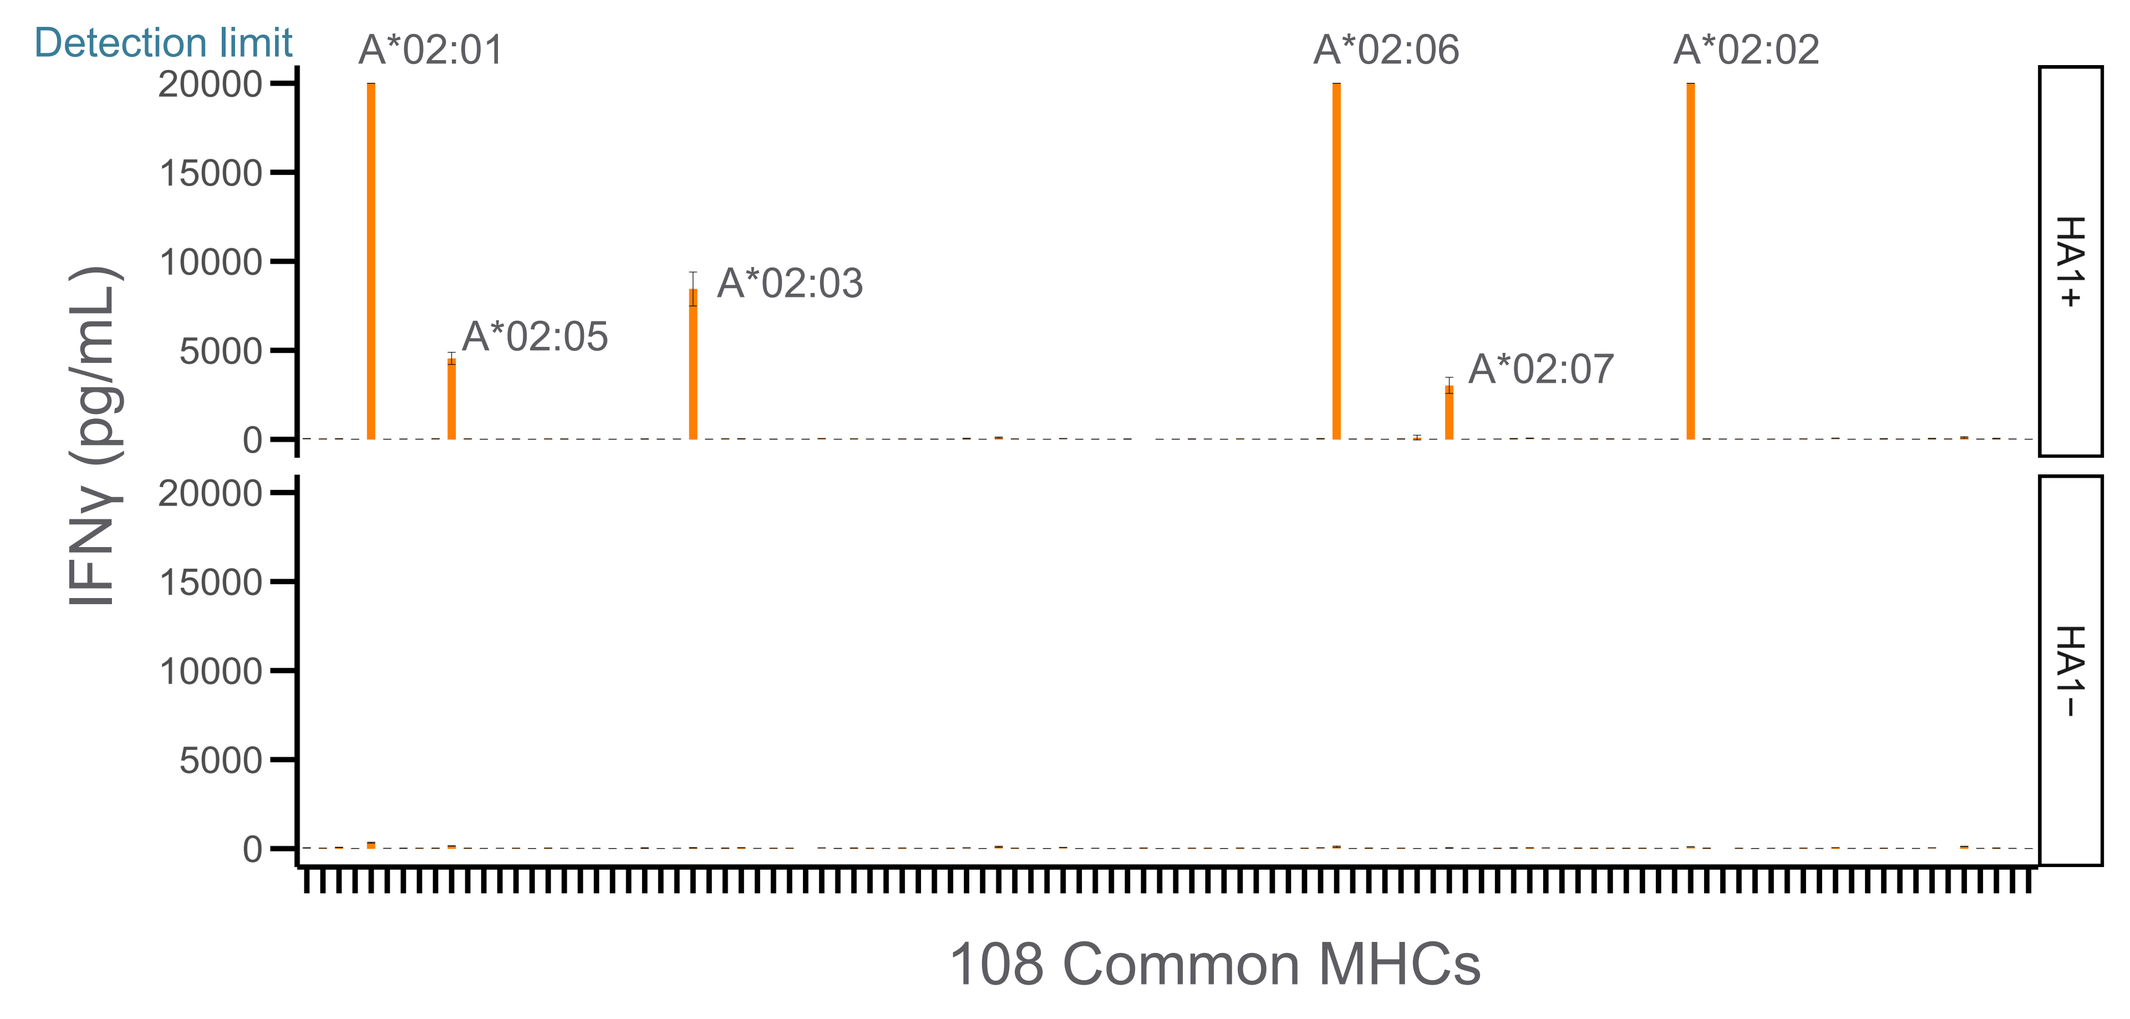
Detection limit
A*02:01
A*02:05
A*02:03
A*02:06
A*02:07
A*02:02
IFNγ (pg/mL)
HA1+
HA1−
108 Common MHCs

TSC-100 was tested in an alloreactivity assay. Endogenous MHCs were knocked out of HEK293T cells using CRISPR/Cas9-gRNA ribonucleoprotein delivery and a cell line was prepared that stably expresses a 90-residue protein fragment that includes the HA-1 sequence. Individual MHCs representing the 108 most prevalent HLA types were then introduced into both the HA-1+ and HA-1- cell lines by lentiviral transduction. Cells in 96-well format were incubated overnight with human primary CD8 T cells expressing TSC-100 and IFN-γ concentration in the supernatant was measured by ELISA. Concentrations above 20,000 pg/mL exceeded the linear detection range of the assay.

TScan's genome-wide screen shows TSC-100 has no detectable off-targets

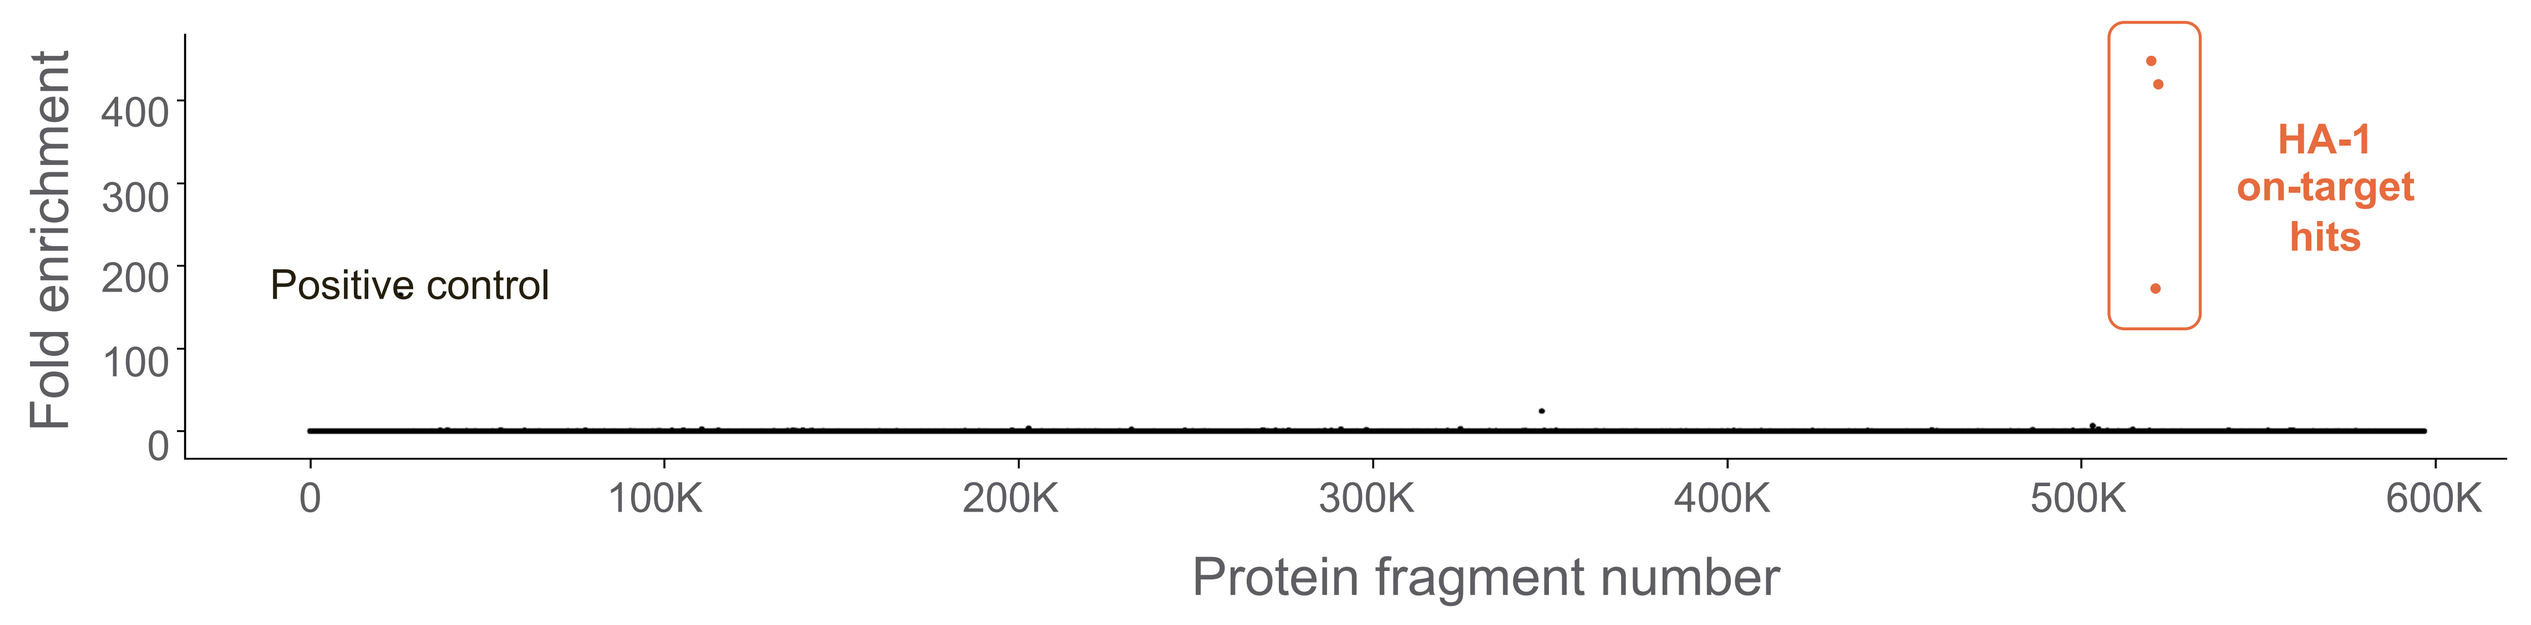
Fold enrichment
Positive control
HA-1 on-target hits
Protein fragment number

TSC-100 was evaluated for off-target reactivity using T-Scan (7). In this genome-wide safety screen, T cells expressing TSC-100 were co-cultured with a library of HLA A*02:01+ target cells, each expressing a different 90-amino acid protein fragment. Collectively, the library includes fragments that tile across every protein in 22-amino acid steps. Fragments are processed naturally by the target cells and the resulting peptides are displayed on cell-surface MHCs. If a T cell recognizes its target, it attempts to kill the target cell, thereby activating a fluorescent reporter. By isolating fluorescent cells and sequencing their expression cassettes, the natural target(s) of the TCR are revealed. Out of ~600,000 clones, only the three clones in our library that contain the HA-1 epitope were significantly enriched in the screen.

Conclusion

329 HA-1-specific TCRs were identified from six HA-1-negative healthy donors using a novel TCR discovery platform. High-throughput screens and in-depth functional assays were then used to identify the most active TCR, which was termed TSC-100. In side-by-side assays, TSC-100 showed superior activity to a clinical-stage HA-1-specific TCR when co-cultured with cell lines expressing low levels of HA-1 and MHC-I. TSC-100 demonstrated no off-target effects in a human genome-wide safety screen and no detectable alloreactivity relative to 108 prevalent HLA types. Based on these results, TSC-100 has been advanced for clinical development, with an IND planned for 2021.

References:
1. Pavletic SZ, Kumar S, Mohty M, et al. *Biol Blood Marrow Transplant,* 2010, **16**, 871-890.
2. Miller JS, Warren EH, van den Brink MR, et al. *Biol Blood Marrow Transplant,* 2010, **16**, 565-586.
3. Marijt W. A. E., Heemskerk M. H. M, Kloosterboer F. M. et. al. *Proc Natl Acad Sci USA,* 2003, **100,** 2742-2747.
4. Spierings E., Kim Y., Hendriks M. et.al. *Biol Blood Marrow Transplant,* 2013, **19,** 1244-1253.
5. Bleakley M, Riddell SR. *Immunol Cell Biol,* 2011, **89,** 396-407.
6. Dossa R. G., Cunningham T., Sommermeyer D. et. al. *Blood,* 2018, **131**, 108-120.
7. Kula T, Dezfulian MH, Wang CI, et al. *Cell,* 2019, **178**, 1016-1028.

Ethics: All clinical samples used in the study were collected by STEMCELL Technologies, StemExpress and HemaCare using their IRB approved protocols.