Product characteristics and clinical trial design for T-Plex: Multiplexed, enhanced T cell receptor-engineered T cell therapy for solid tumors

Shrikanta Chattopadhyay, Antoine Boudot, Ribhu Nayar, Hannah Bader, Tary Traore, Nancy Nabilsi, Jenny Tadros, Badr Kiaf, Yifan Wang, Victor Ospina, Alok Das Mohapatra, Zhonghua Zhu, Qikai Xu, Krista Daniels, Briana Zimmerman, Mollie Jurewicz, Amanda Kordosky, Chris Malcuit, Yun Wang, Marlyane Motta, Nina Abelowitz, Jim Murray, Ray Lockard, Cagan Gurer, Debora Barton, Gavin MacBeath

TScan Therapeutics, Waltham, MA

#2554

Background and Rationale

- Most patients fail checkpoint immunotherapy due to lack of sufficient endogenous anti-tumor T cells
- A potential solution is to engineer T cells with exogenous T cell receptors (TCRs) that target tumor antigens
- However solid tumors are notoriously heterogenous with heterogenous target antigen expression
- Solid tumors have also been recently recognized to have HLA loss of heterozygosity (LOH) in up to 40% of tumors
- First generation TCR-Ts targeting single antigens had limited response rates (30-50%) and short durations of response (3-4 months)
- TScan's solution is to develop multiplexed TCR-Ts targeting different antigens on different HLA types
- TCR-T cells also have genetic enhancements to enable potent tumor killing and long-term persistence.

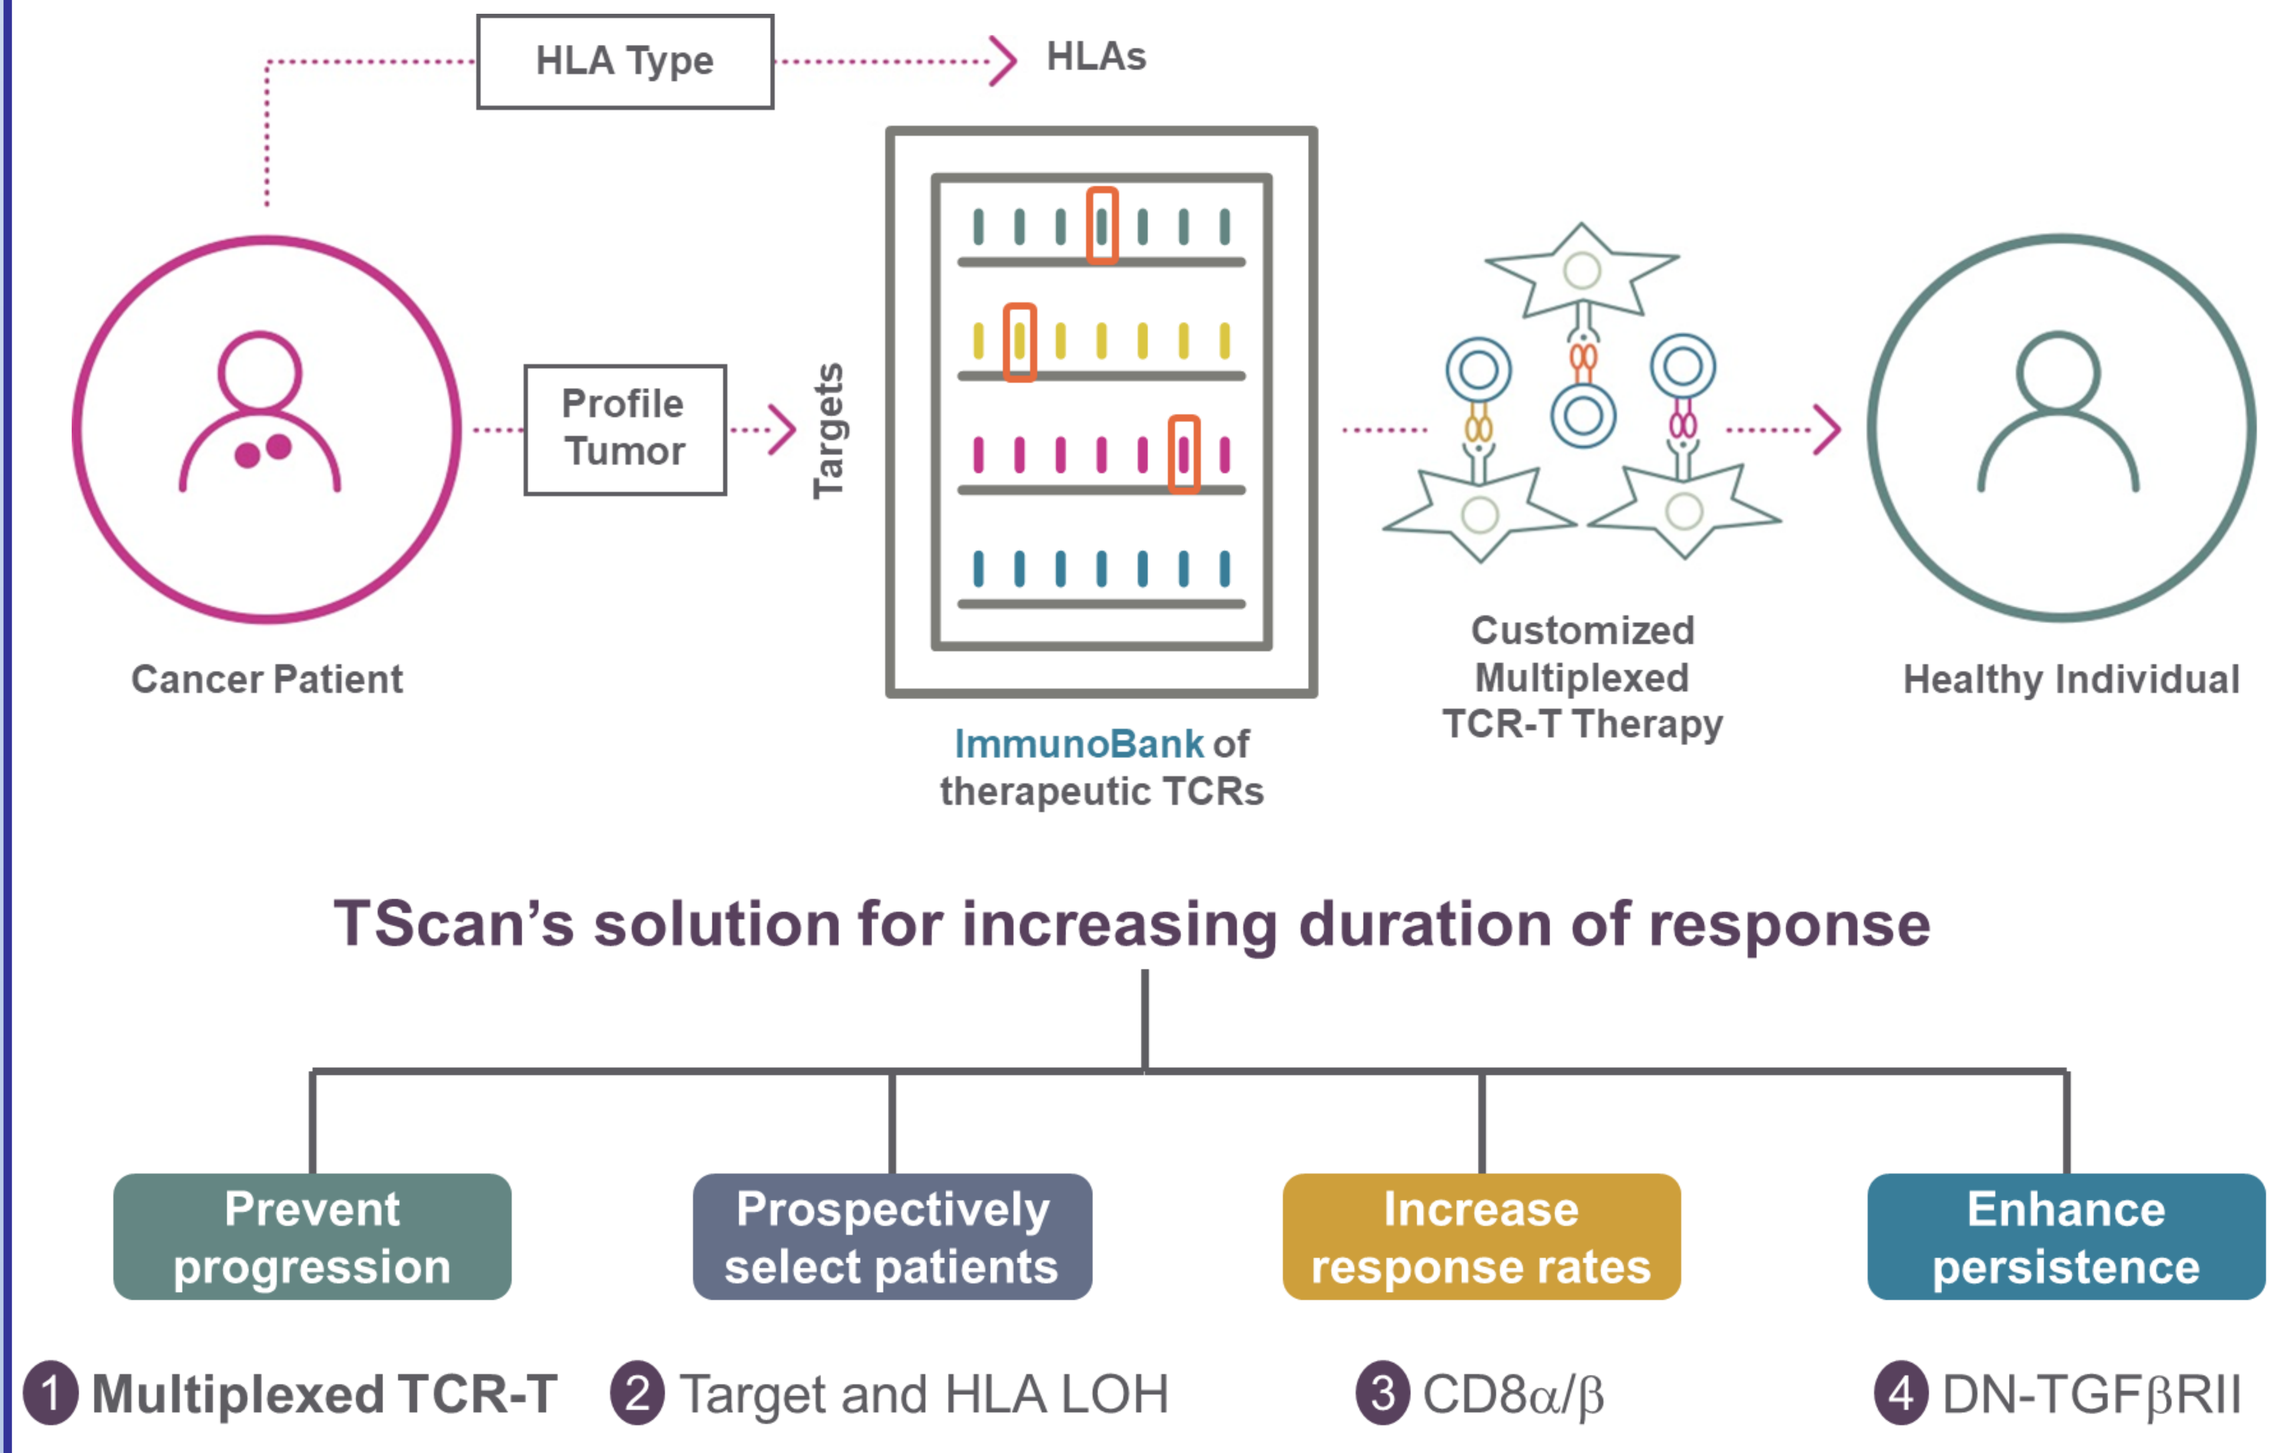
HLA Type
HLAs
Profile Tumor
Targets
Cancer Patient
ImmunoBank of therapeutic TCRs
Customized Multiplexed TCR-T Therapy
Healthy Individual
TScan's solution for increasing duration of response
Prevent progression
Prospectively select patients
Increase response rates
Enhance persistence
1 Multiplexed TCR-T
2 Target and HLA LOH
3 CD8α/β
4 DN-TGFβRII

Target heterogeneity in solid tumors

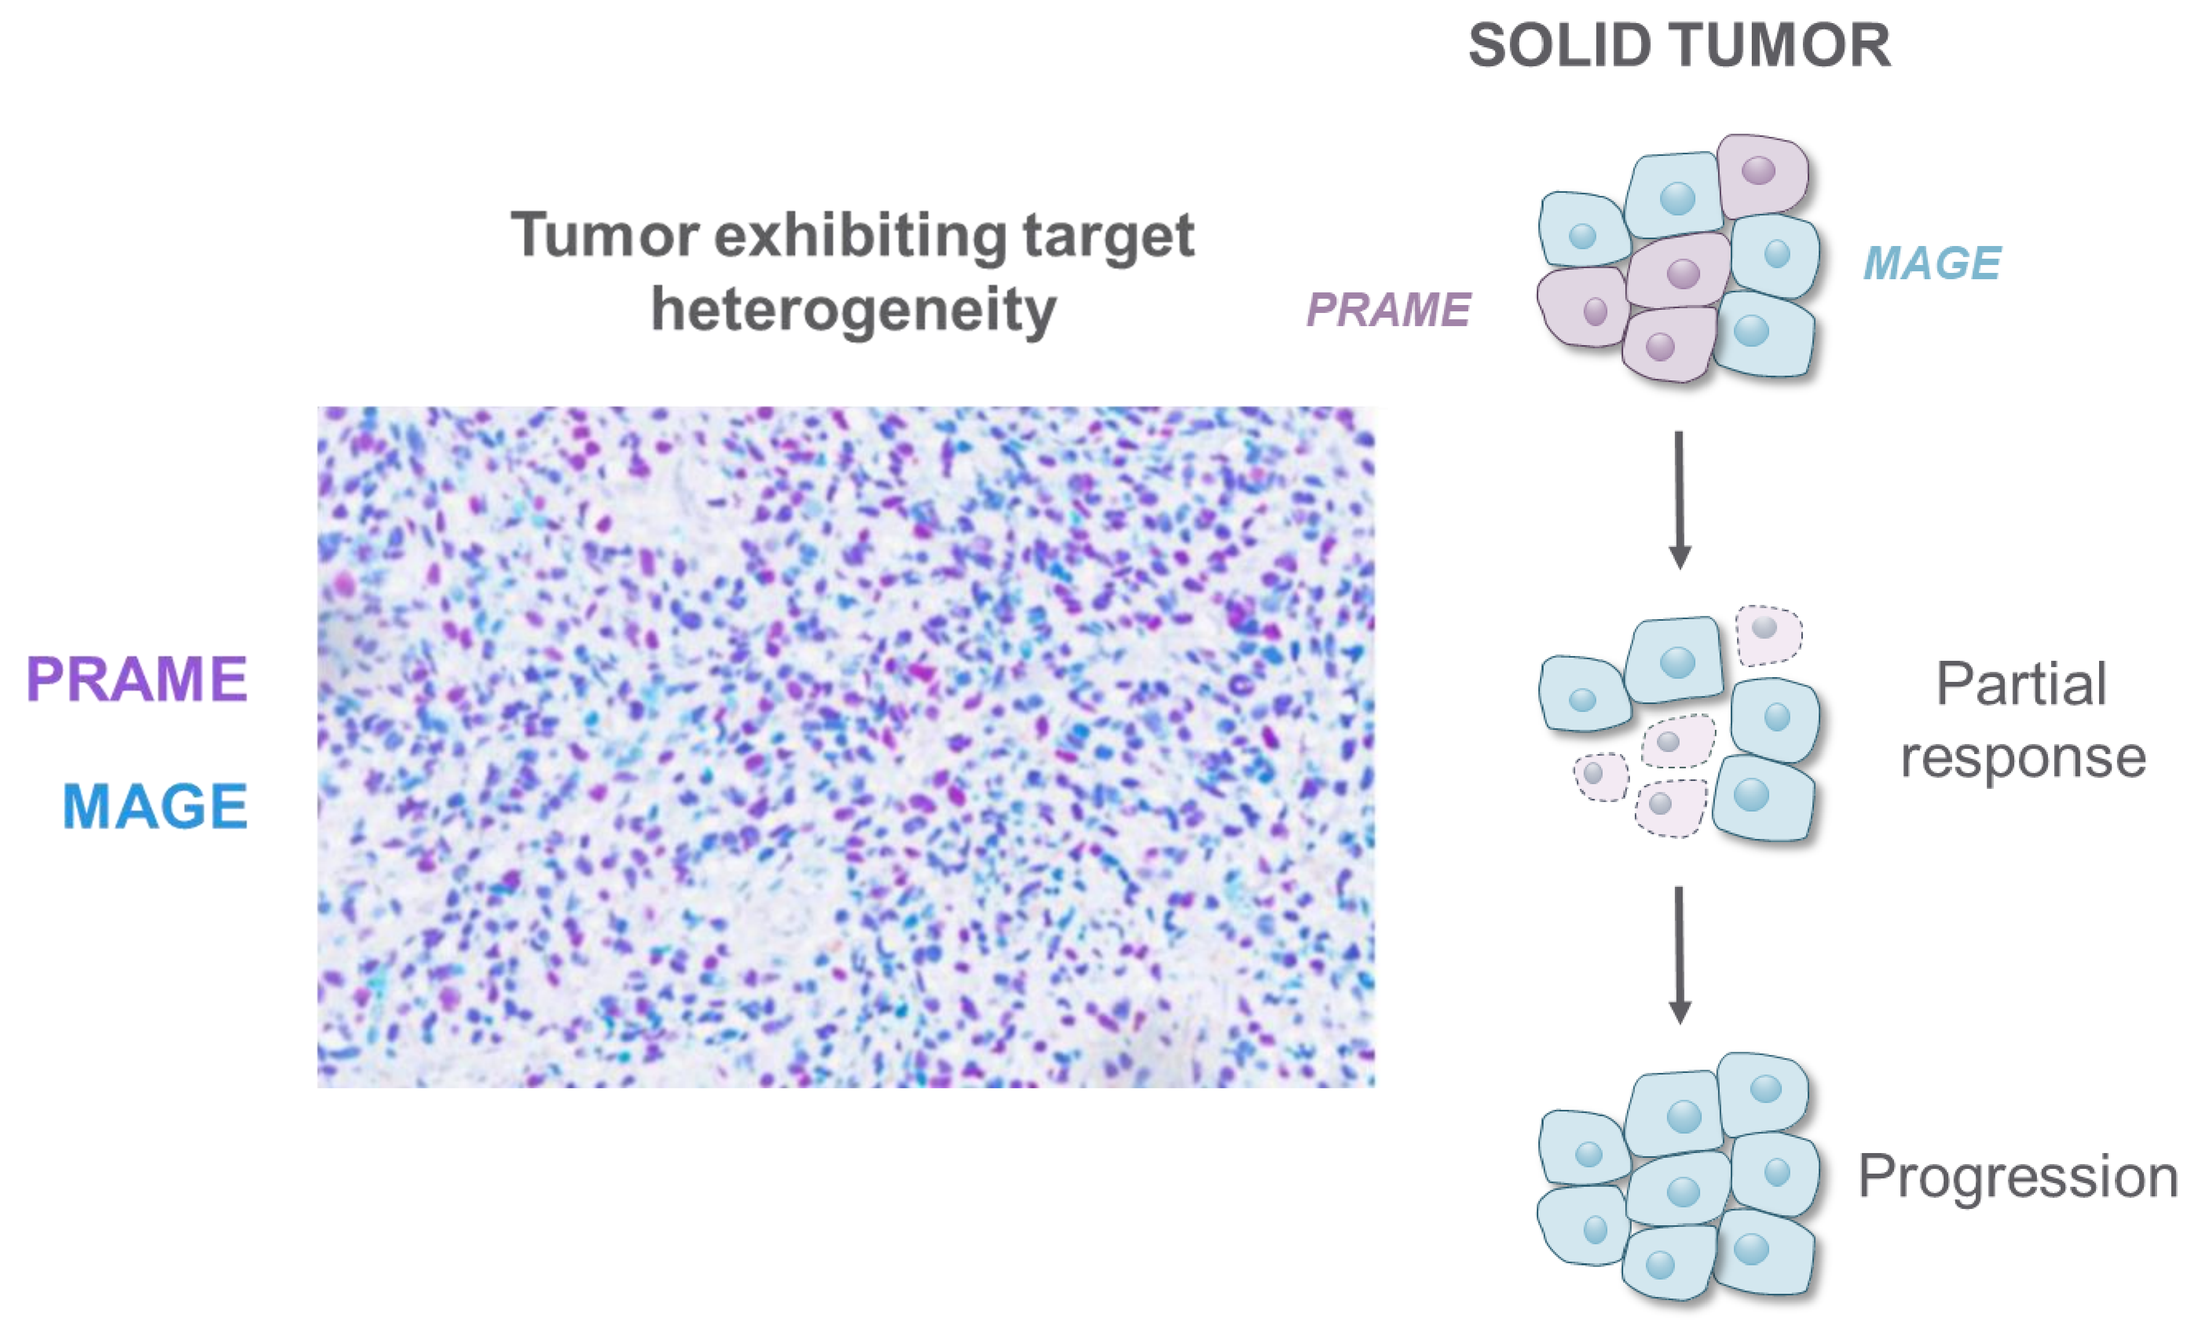
SOLID TUMOR
Tumor exhibiting target heterogeneity
PRAME
MAGE
PRAME
MAGE
Partial response
Progression

- Melanoma samples were stained with PRAME (purple) and MAGE-C2 antibodies (blue).
- Tumor cells were noted to be positive for either PRAME or MAGE-C2 or both
- Targeting single antigens is expected to result in partial responses in these tumors with rapid progression

HLA loss of heterozygosity (LOH) is prevalent and overlooked in solid tumors

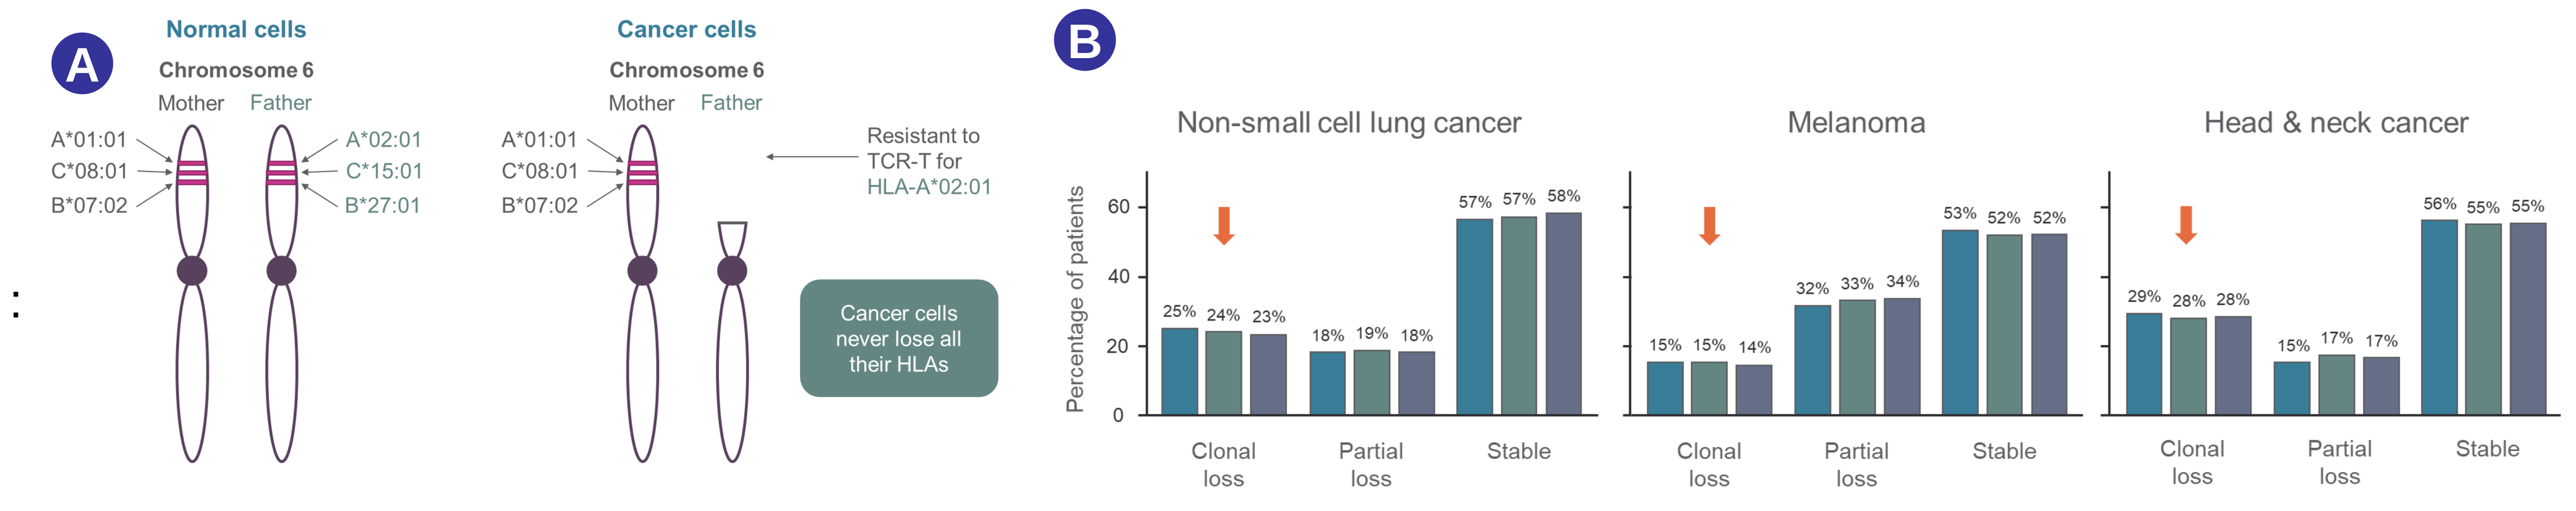
A
Normal cells
Chromosome 6
Mother Father
A*01:01
C*08:01
B*07:02
A*02:01
C*15:01
B*27:01
Cancer cells
Chromosome 6
Mother Father
A*01:01
C*08:01
B*07:02
Resistant to TCR-T for HLA-A*02:01
Cancer cells never lose all their HLAs
B
Non-small cell lung cancer
Melanoma
Head & neck cancer
Percentage of patients
25% 24% 23%
18% 19% 18%
57% 57% 58%
15% 15% 14%
32% 33% 34%
53% 52% 52%
29% 28% 28%
15% 17% 17%
56% 55% 55%
Clonal loss
Partial loss
Stable

(**A**) HLA loss of heterozygosity (LOH) generally occurs through loss of one HLA haplotype on chromosome 6. (**B**) Novel HLA LOH detection algorithm using tumor/ normal comparisons of Tempus tumor data identifies HLA clonal loss in 15-30% of common solid tumors. Those tumors cannot respond to single-targeted TCR-Ts.

Multiplexing and TCR-T selection overcome heterogeneity and HLA LOH

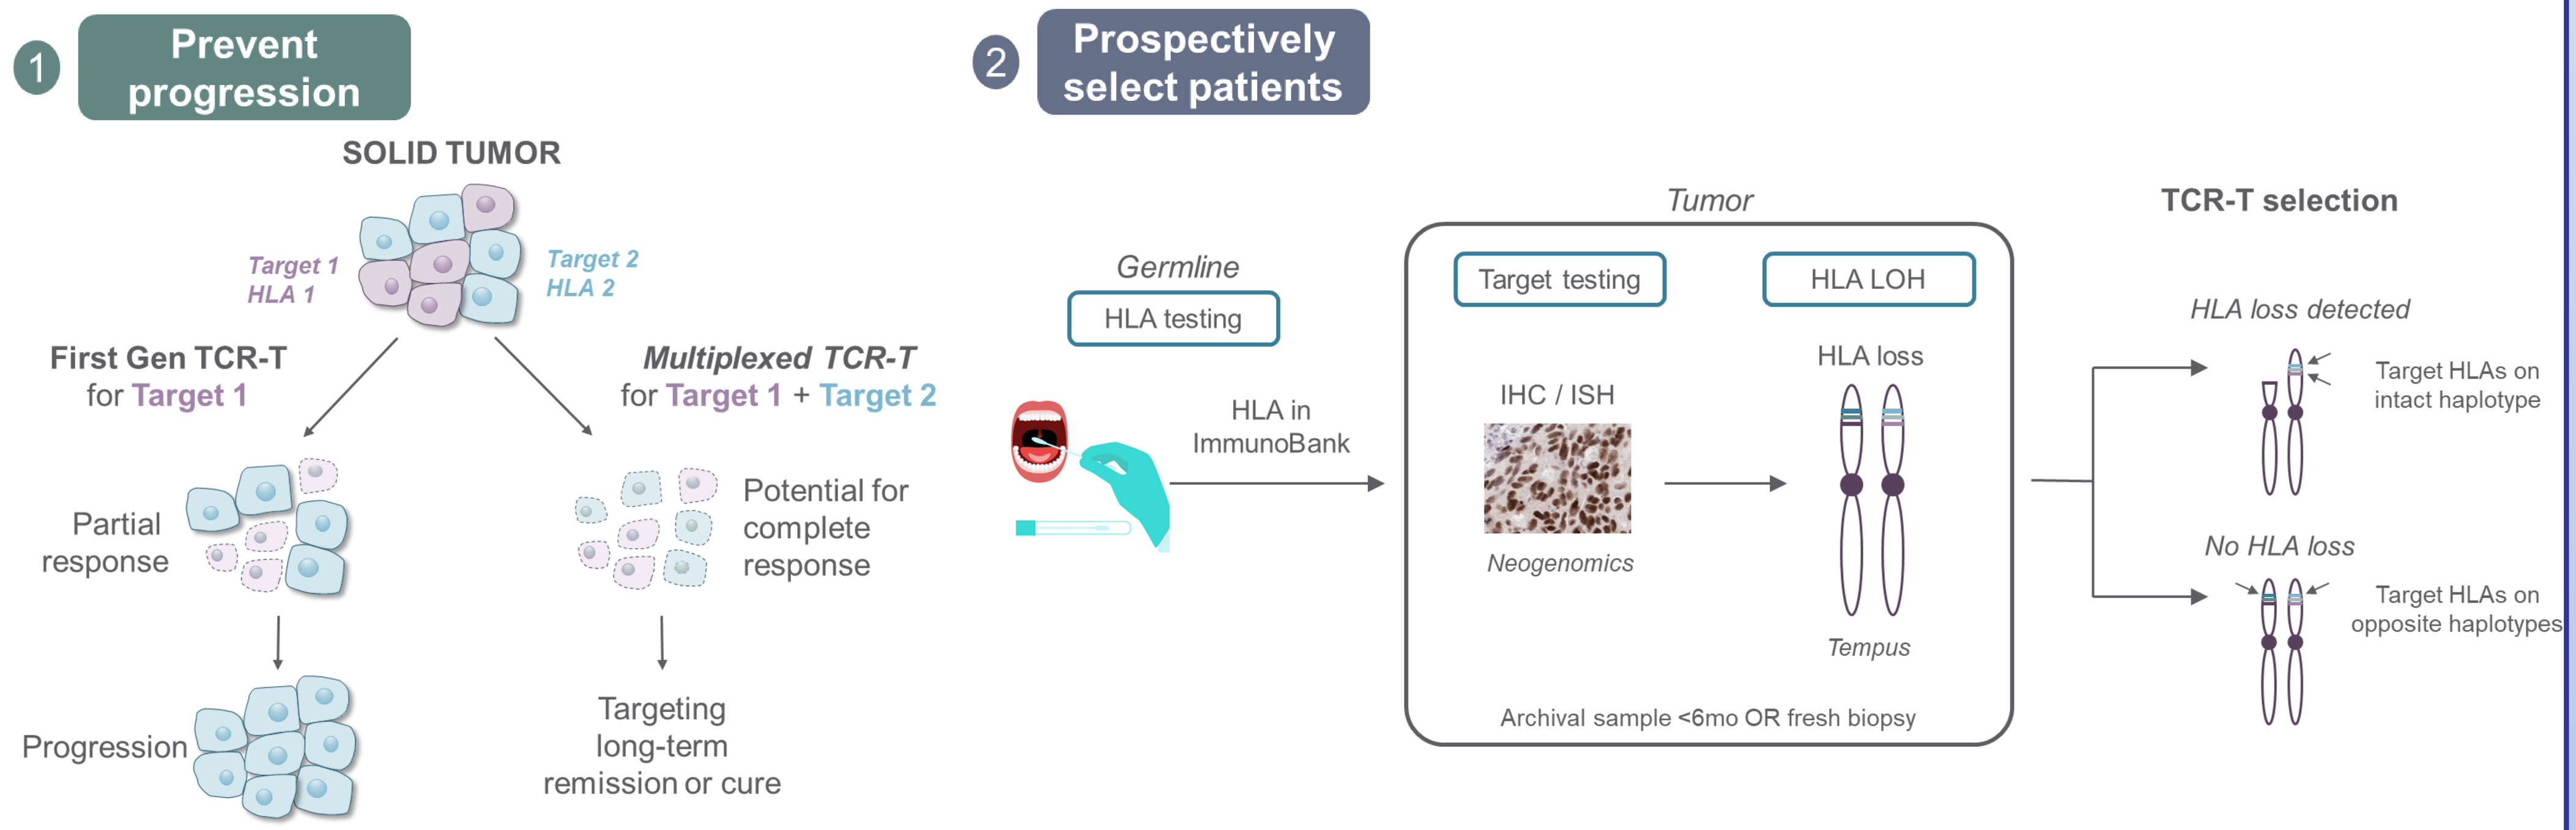
1 Prevent progression
2 Prospectively select patients
SOLID TUMOR
Target 1 HLA 1
Target 2 HLA 2
First Gen TCR-T for Target 1
Multiplexed TCR-T for Target 1 + Target 2
Partial response
Potential for complete response
Progression
Targeting long-term remission or cure
Germline
HLA testing
HLA in ImmunoBank
Tumor
Target testing
HLA LOH
IHC / ISH
Neogenomics
HLA loss
Tempus
Archival sample <6mo OR fresh biopsy
TCR-T selection
HLA loss detected
Target HLAs on intact haplotype
No HLA loss
Target HLAs on opposite haplotypes

(**1**) **Prevent Progression**: First generation TCR-Ts targeted single antigens on single HLA types in partial responses and rapid progression. Multiplexing TCR-Ts targeting different target antigens on different HLA types has the potential for complete responses and long-term remissions (**2**) **Prospectively select patients**: Germline HLA typing is followed by testing tumors for target antigens and HLA LOH. TCR-T selection can be used to overcome HLA LOH.

TCR-T cell function is enhanced with CD8α/β coreceptors and DN-TGFβRII

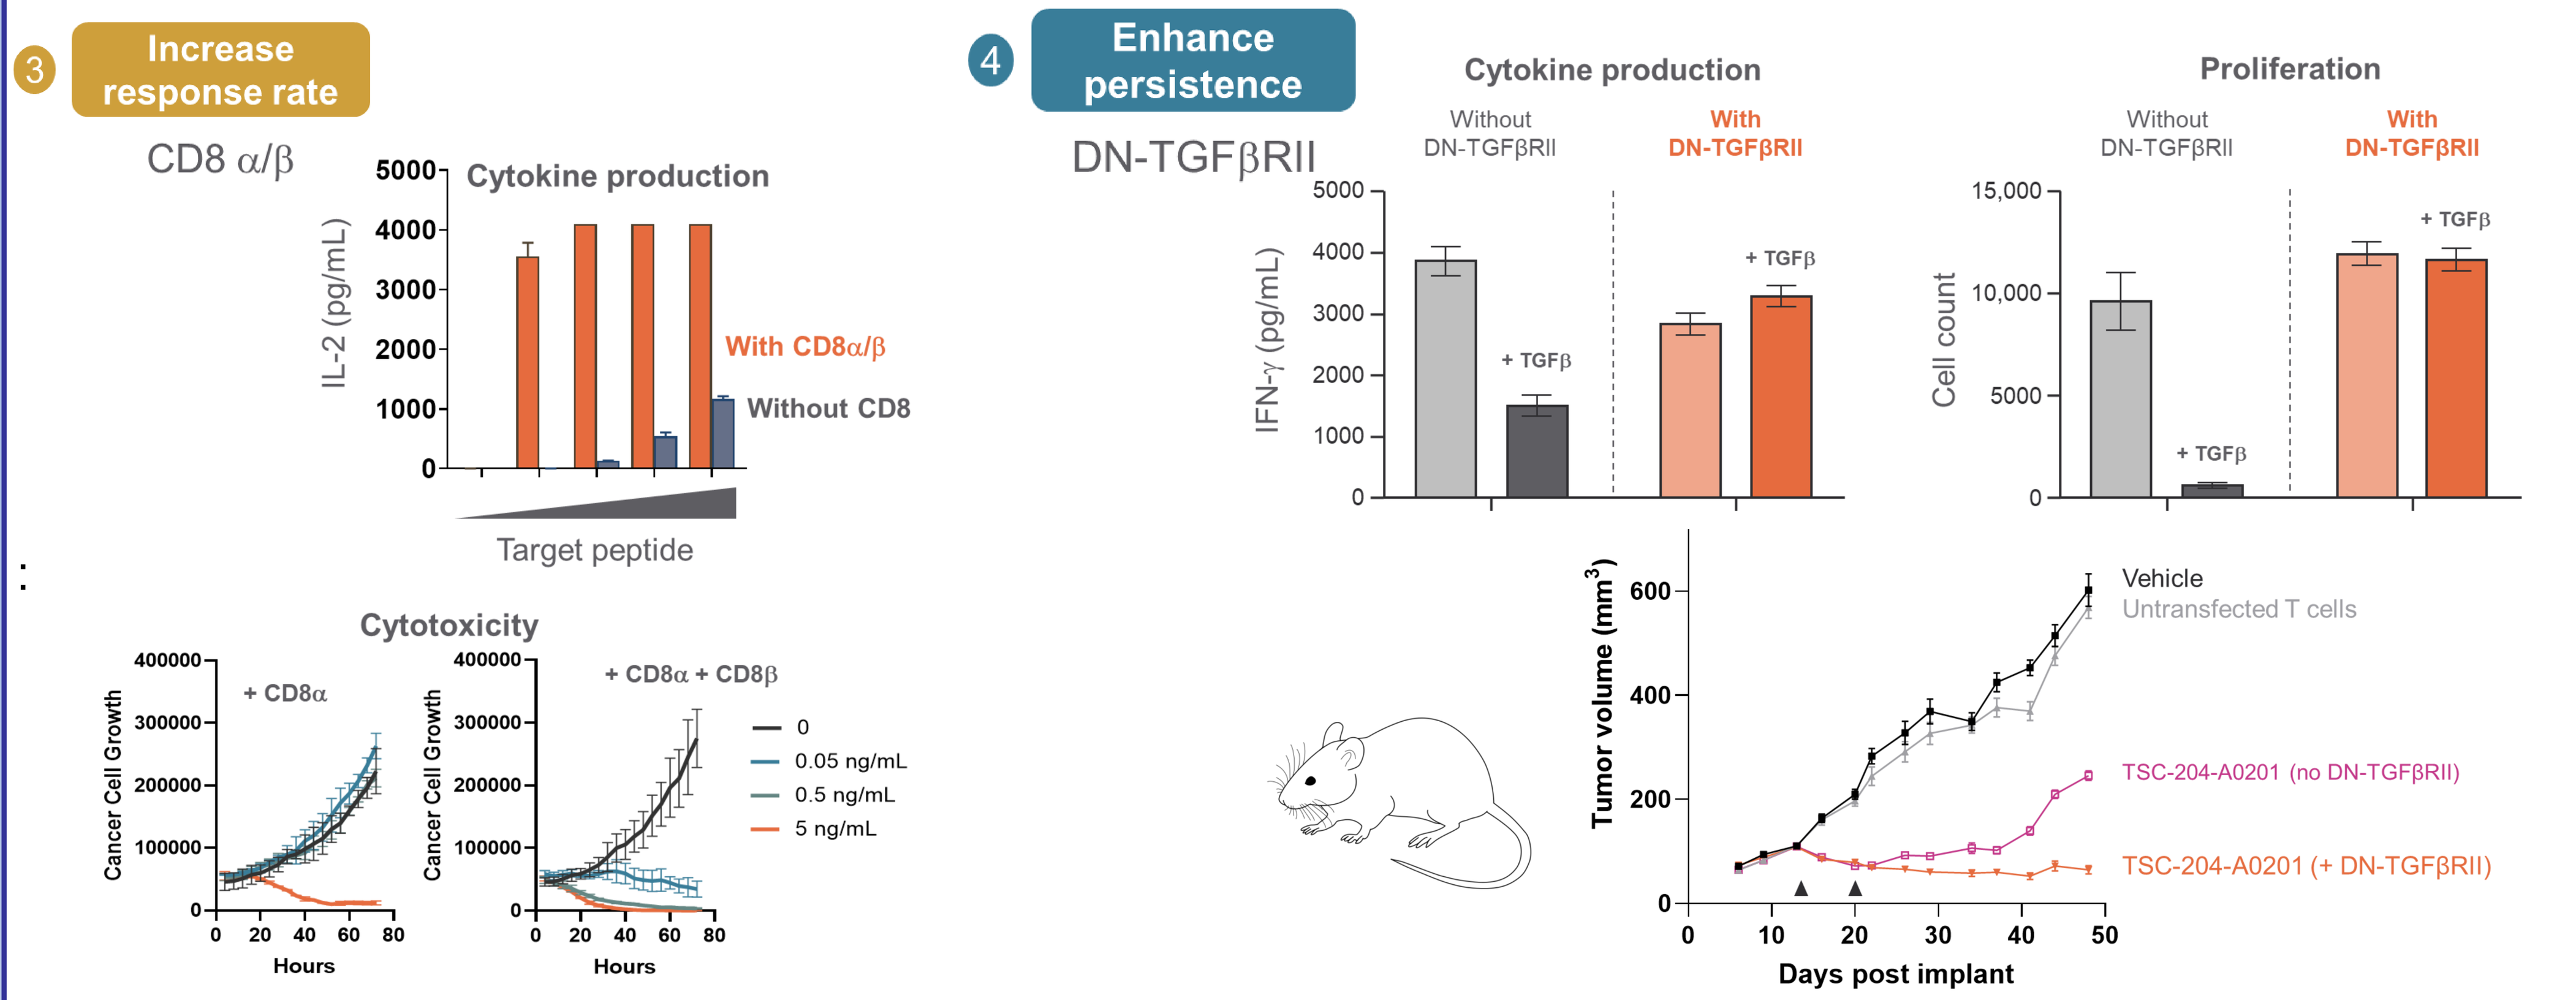
3 Increase response rate
CD8 α/β
Cytokine production
IL-2 (pg/mL)
With CD8α/β
Without CD8
Target peptide
Cytotoxicity
+ CD8α
+ CD8α + CD8β
Cancer Cell Growth
Hours
0
0.05 ng/mL
0.5 ng/mL
5 ng/mL
4 Enhance persistence
DN-TGFβRII
Cytokine production
Without DN-TGFβRII
With DN-TGFβRII
IFN-γ (pg/mL)
+ TGFβ
Proliferation
Cell count
Tumor volume (mm3)
Days post implant
Vehicle
Untransfected T cells
TSC-204-A0201 (no DN-TGFβRII)
TSC-204-A0201 (+ DN-TGFβRII)

(3) Increase response rate: CD4+ T cells engineered to express TCR+CD8α+CD8β coreceptors had ~100-fold higher cytokine production and cytotoxicity versus CD4+ cells expressing TCR alone or TCR+CD8α. (4) **Enhance persistence**: Expression of dominant-negative TGFβRII (DN-TGFβRII) results in ~2-fold higher cytokine production and ~10-fold higher T-cell expansion in the presence of TGFβ *in vitro* and durable responses of tumors *in vivo*.

Screening protocol pre-identifies patients eligible for treatment protocol

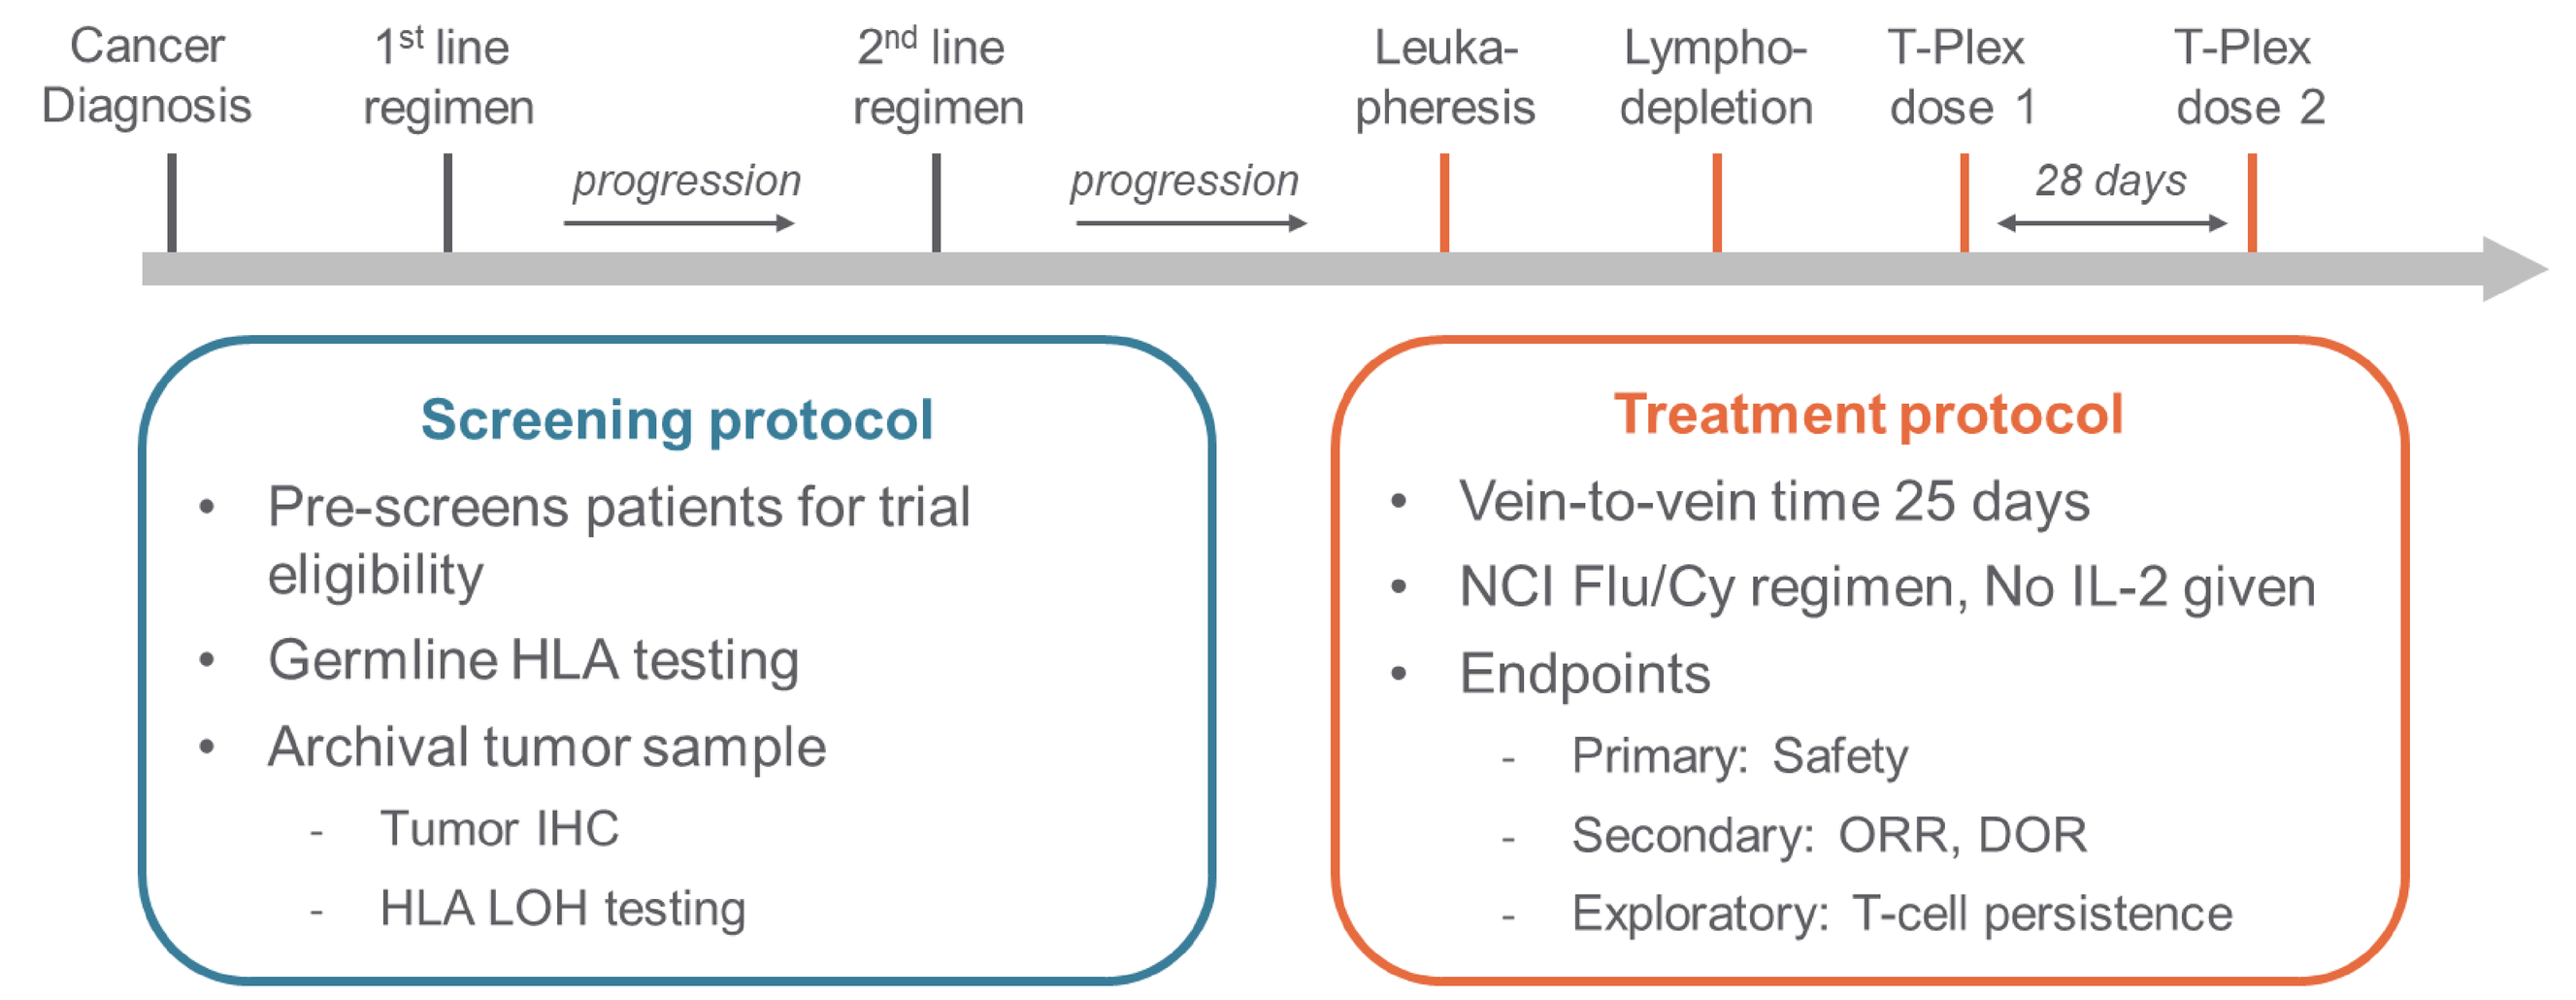
Cancer Diagnosis
1st line regimen
progression
2nd line regimen
progression
Leuka-pheresis
Lympho-depletion
T-Plex dose 1
T-Plex dose 2
28 days
Screening protocol
Pre-screens patients for trial eligibility
Germline HLA testing
Archival tumor sample
Tumor IHC
HLA LOH testing
Treatment protocol
Vein-to-vein time 25 days
NCI Flu/Cy regimen, No IL-2 given
Endpoints
Primary: Safety
Secondary: ORR, DOR
Exploratory: T-cell persistence

Patients with melanoma, non-small cell lung cancer, head and neck cancer, cervical cancer, ovarian cancer or anogenital cancers are eligible. Screening includes germline HLA typing then archival tumor testing for antigens and HLA LOH any time during standard cancer treatment. Treatment involves 1-2 doses of TCR-T cell therapy after lymphodepletion.

Dose escalation scheme provides rapid path to multiplexing from dose level 3

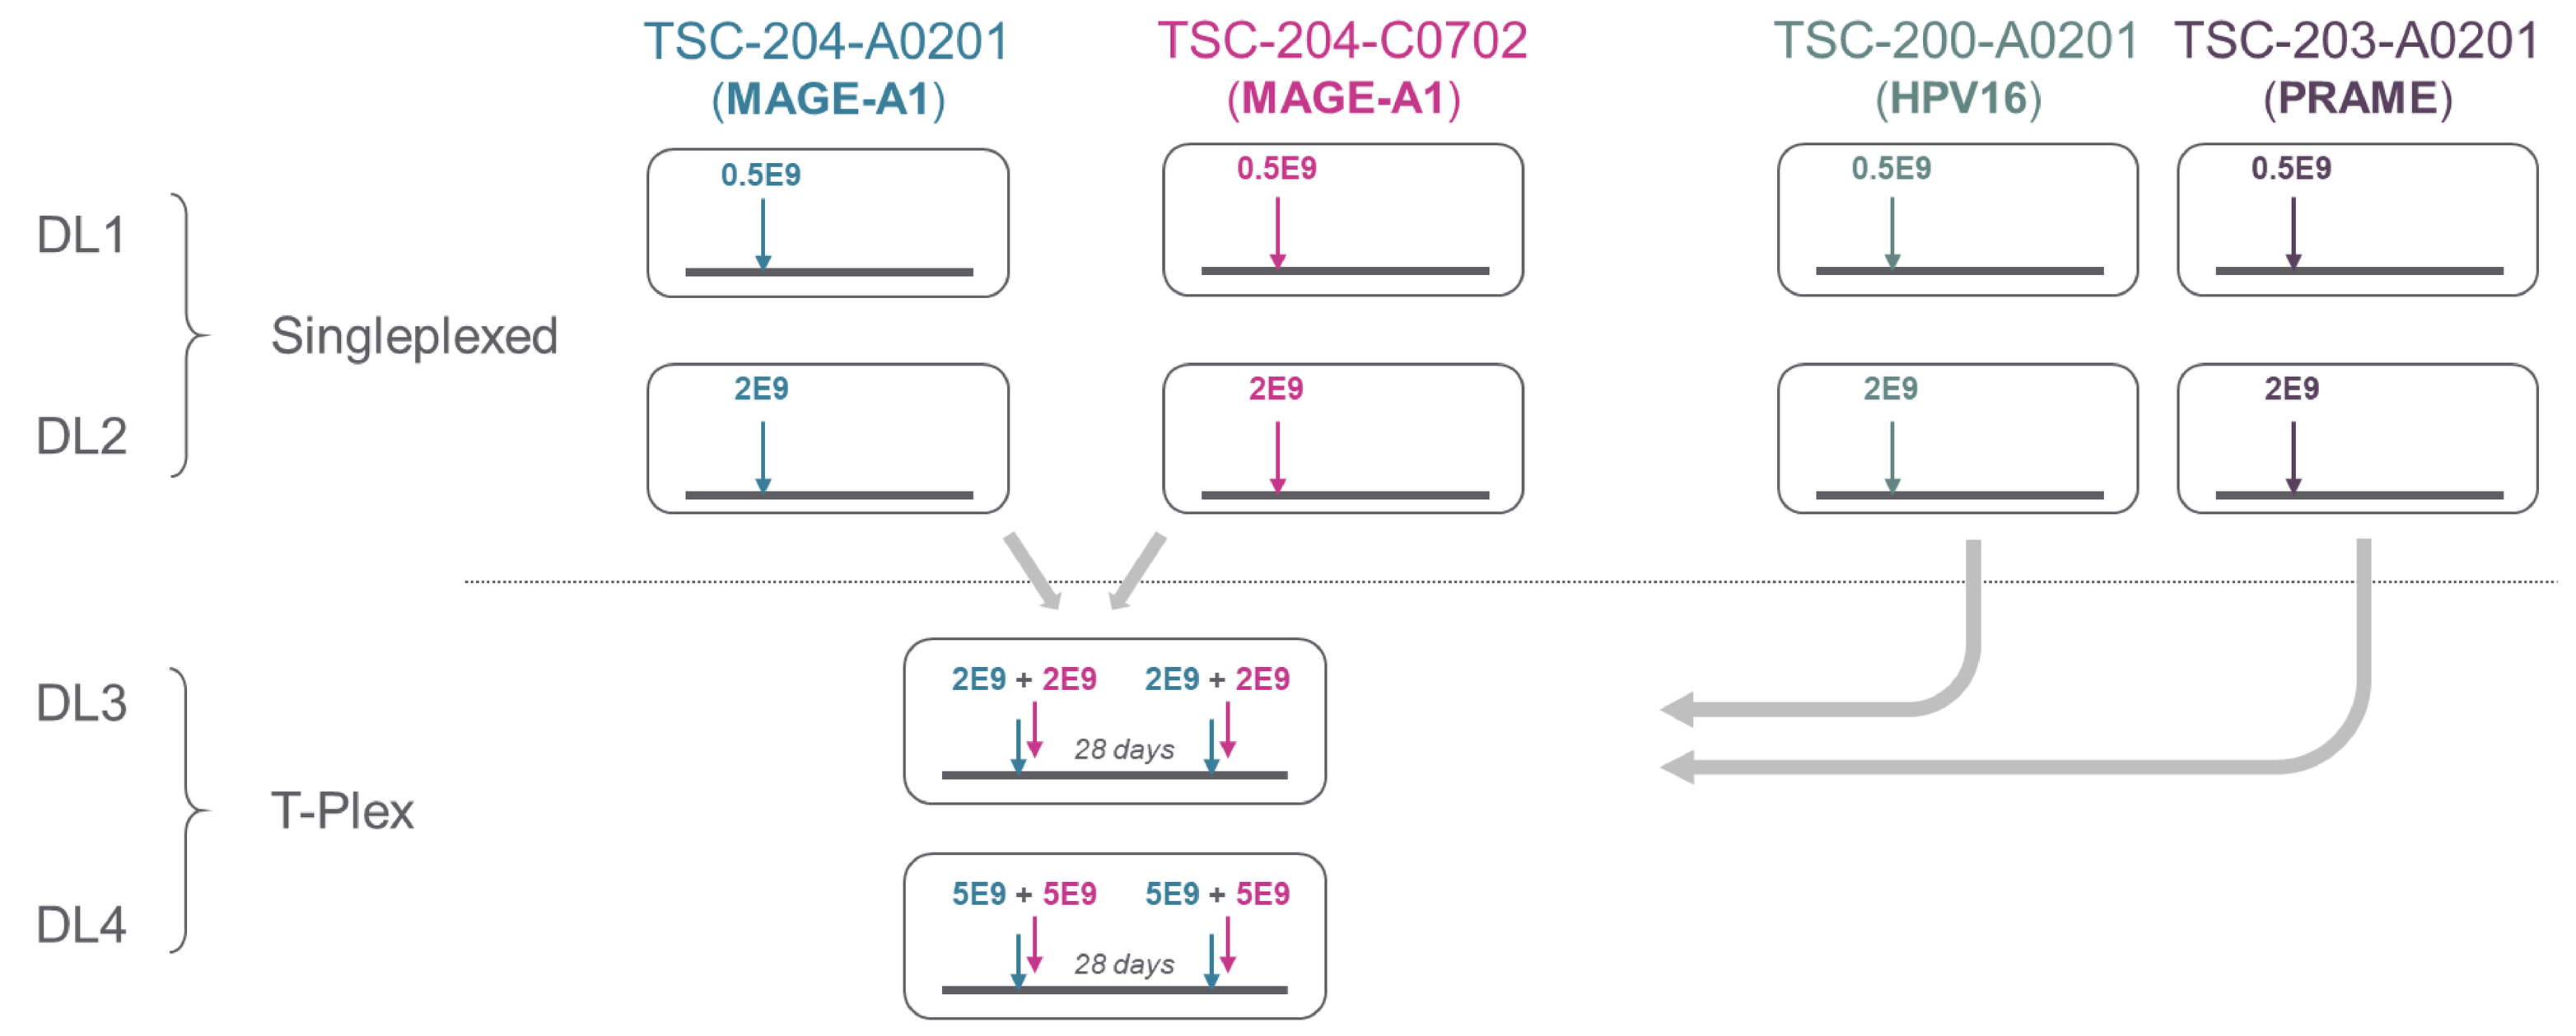
TSC-204-A0201 (MAGE-A1)
TSC-204-C0702 (MAGE-A1)
TSC-200-A0201 (HPV16)
TSC-203-A0201 (PRAME)
DL1
DL2
Singleplexed
0.5E9
2E9
DL3
DL4
T-Plex
2E9 + 2E9
5E9 + 5E9
28 days

INDs have been cleared for TCR-Ts targeting MAGE-A1 on HLA-A*02:01 (TSC-204-A0201), HLA-C*07:02 (TSC-204-C0702) and their combination. The FDA cleared protocol allows multiplexing and repeat dosing from dose level 3. As additional INDs for TCR-Ts targeting HPV16 and PRAME are cleared, they will follow the same dose escalation scheme.

Eligibility for multiplexed therapy increases with growing collection of TCR-Ts

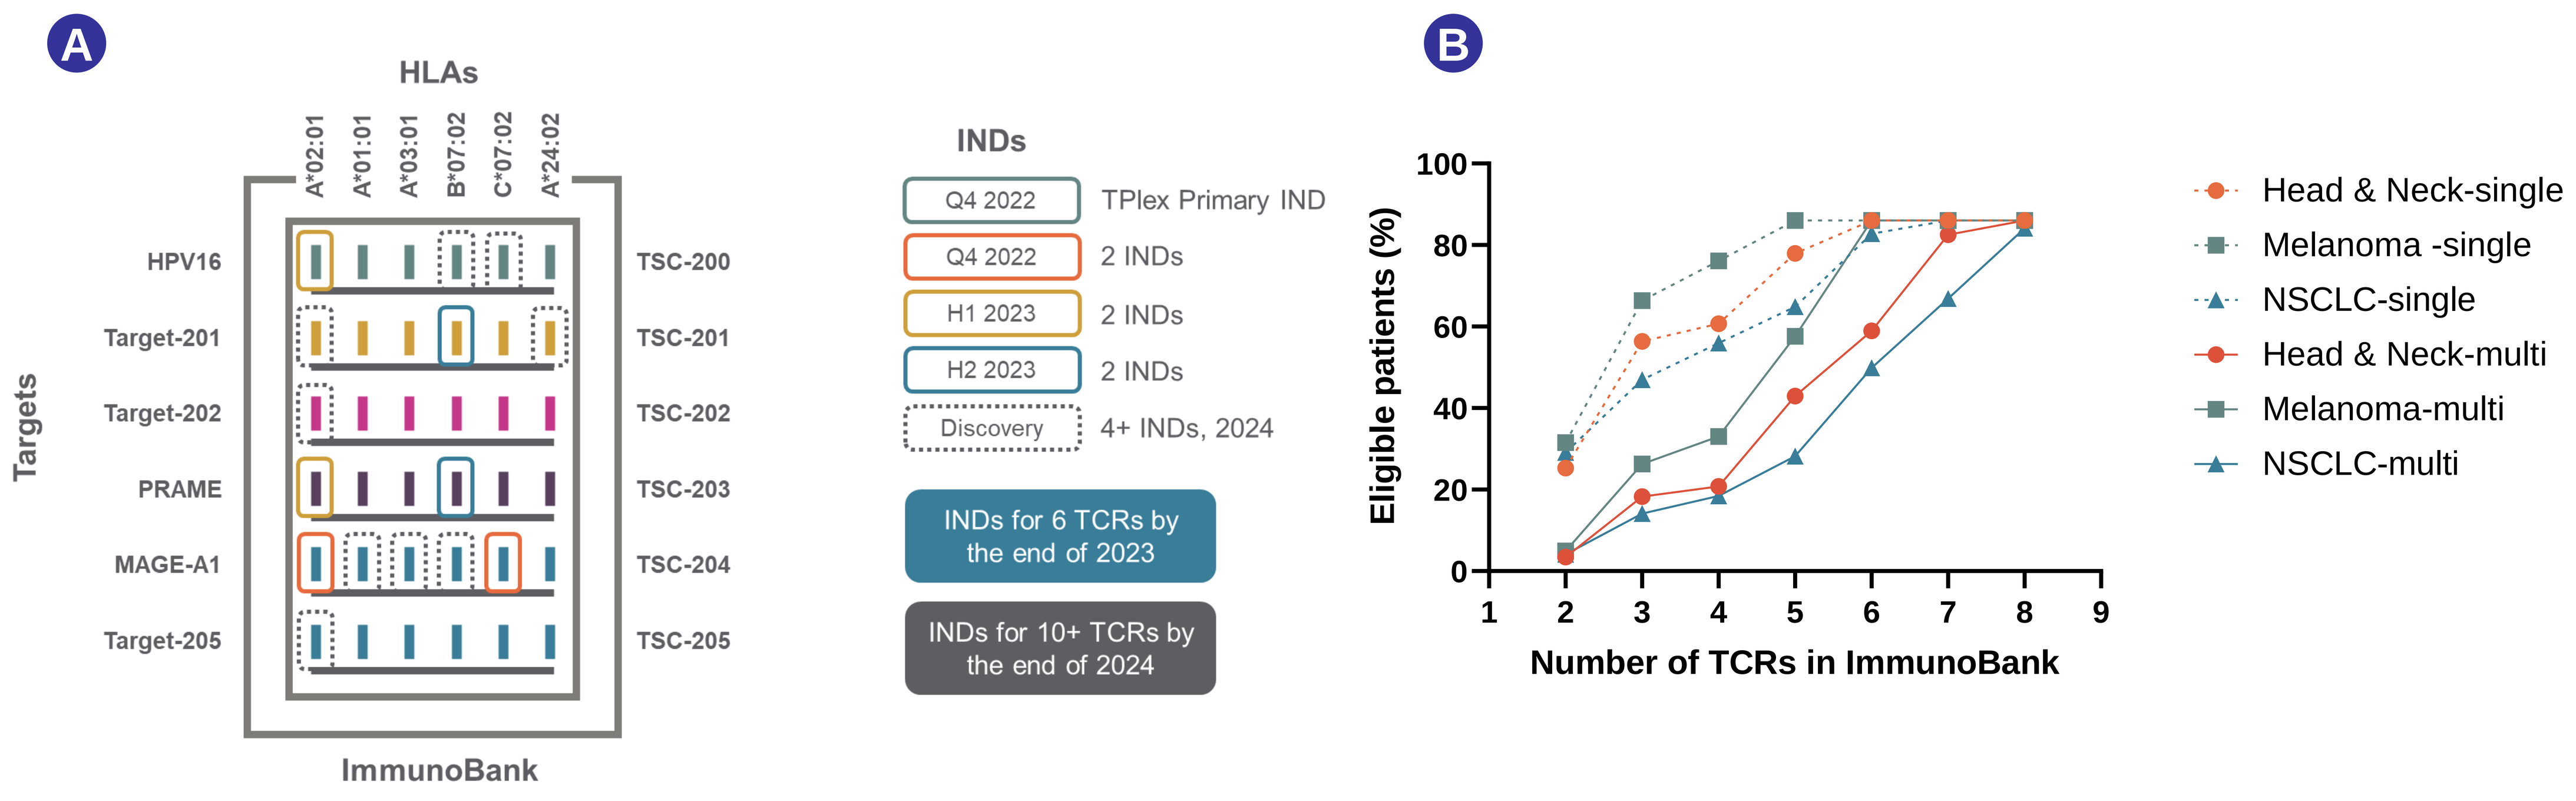
A
HLAs
A*02:01
A*01:01
A*03:01
B*07:02
C*07:02
A*24:02
Targets
HPV16
Target-201
Target-202
PRAME
MAGE-A1
Target-205
TSC-200
TSC-201
TSC-202
TSC-203
TSC-204
TSC-205
ImmunoBank
INDs
Q4 2022 TPlex Primary IND
Q4 2022 2 INDs
H1 2023 2 INDs
H2 2023 2 INDs
Discovery 4+ INDs, 2024
INDs for 6 TCRs by the end of 2023
INDs for 10+ TCRs by the end of 2024
B
Eligible patients (%)
Number of TCRs in ImmunoBank
Head & Neck-single
Melanoma -single
NSCLC-single
Head & Neck-multi
Melanoma-multi
NSCLC-multi

(**A**) The ImmunoBank is the collection of TCR-Ts from which 1-2 therapies for individual patients are chosen. INDs for two TCR-Ts and the TPlex combination have been cleared. Two additional INDs are on track to be cleared by mid-2023 and 6 TCR-Ts are expected to be available by end of 2023. (**B**) As the number of TCR-T choices grows, the number of solid tumor patients eligible for singleplexed therapy (dotted lines) or multiplexed therapy (solid lines) increases.